PHARMACODYNAMIC EFFECTS OF THE S2R MODULATOR CT1812 IN ALZHEIMER'S DISEASE (AD) PATIENTS OBSERVED IN A META-ANALYSIS OF CSF PROTEOMES FROM SPARC AND SHINE PART A

K. Pandey[3], D. Duong[1], L. Waybright[2], B. Lizama[2], A. Mecca[4], C. van Dyck[5], M. Grundman[2], K. Blennow[6], H. Zetterberg[6], A.O. Caggiano[2], N. Seyfried[1], Mary E Hamby[2]

Affiliations: (1)Emory University School of Medicine, Biochemistry, Atlanta, GA, United States of America, (2)Cognition Therapeutics, Research, Pittsburgh, PA, United States of America, (3)Emtherapro Inc, Systems Biology, Atlanta, GA, United States of America, (4)Yale University, New Haven, CT, USA, (5)Yale School of Medicine, New Haven, CT, USA, (6) Clinical Neurochemistry Lab, Institute of Neuroscience and Physiology, Sahlgrenska Academy, University of Gothenburg, Mölndal, Sweden

BACKGROUND

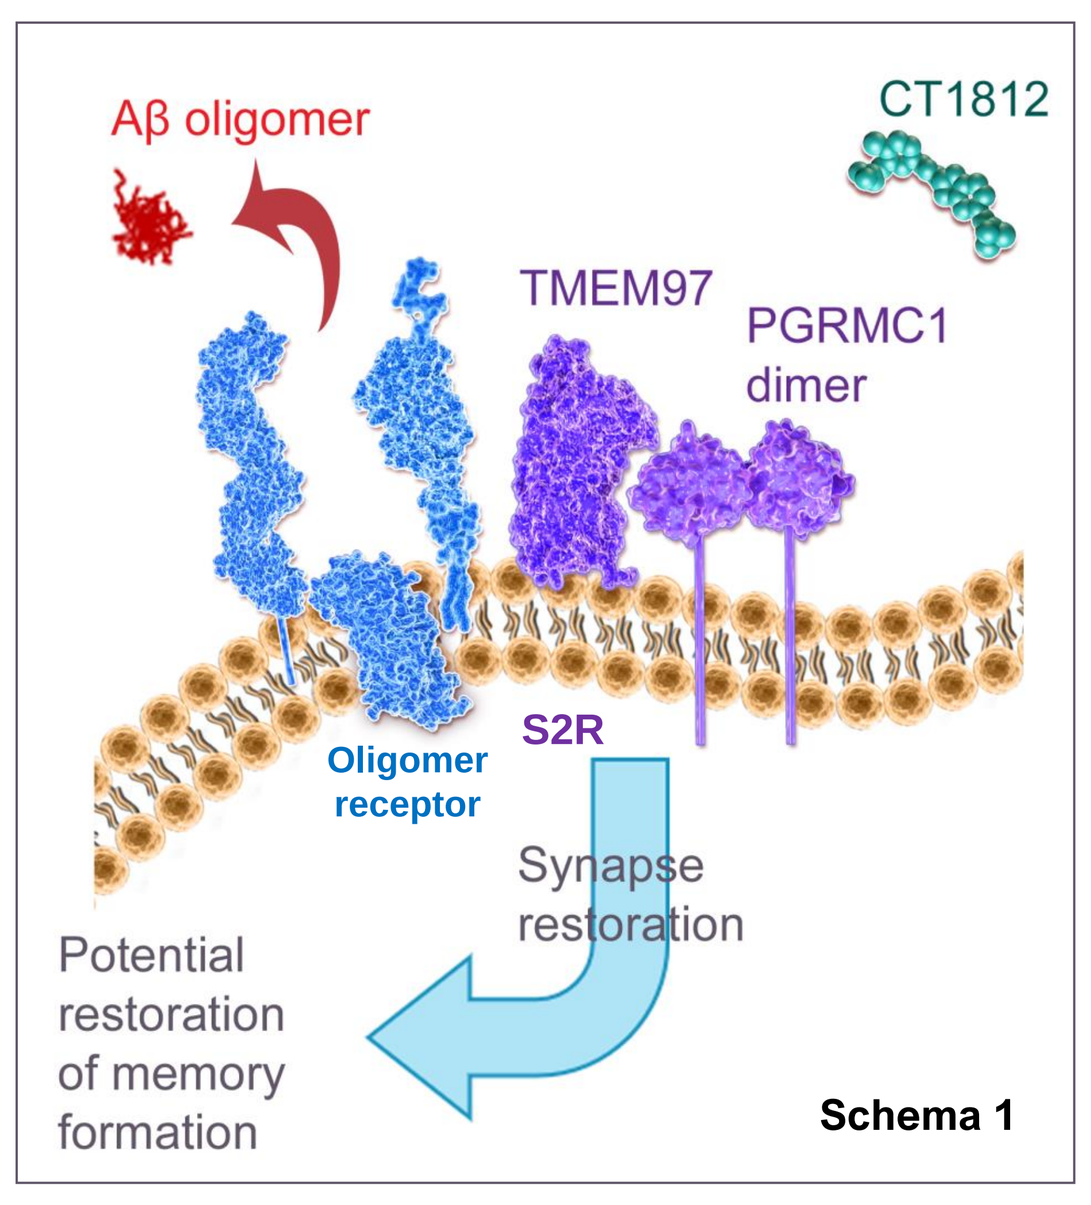
Aβ oligomer
CT1812
TMEM97
PGRMC1 dimer
S2R
Oligomer receptor
Synapse restoration
Potential restoration of memory formation
Schema 1

The sigma-2 receptor (S2R) is encoded by TMEM97, a four-domain transmembrane protein that forms a complex with progesterone receptor membrane component 1 (PGRMC1). CT1812 is a highly brain-penetrant small molecule modulator of S2R, that displaces Aβ oligomers bound to neuronal synapses[1] (**Schema 1**). In preclinical studies, CT1812 protects synapses, facilitates their restoration and improves cognitive performance in transgenic Alzheimer's disease (AD) mice[1]. CT1812 is in clinical development for AD. SHINE (NCT03507790), ongoing, and SPARC (NCT03493282), now completed, are randomized double-blind placebo-controlled trials **(Schema 2)**.

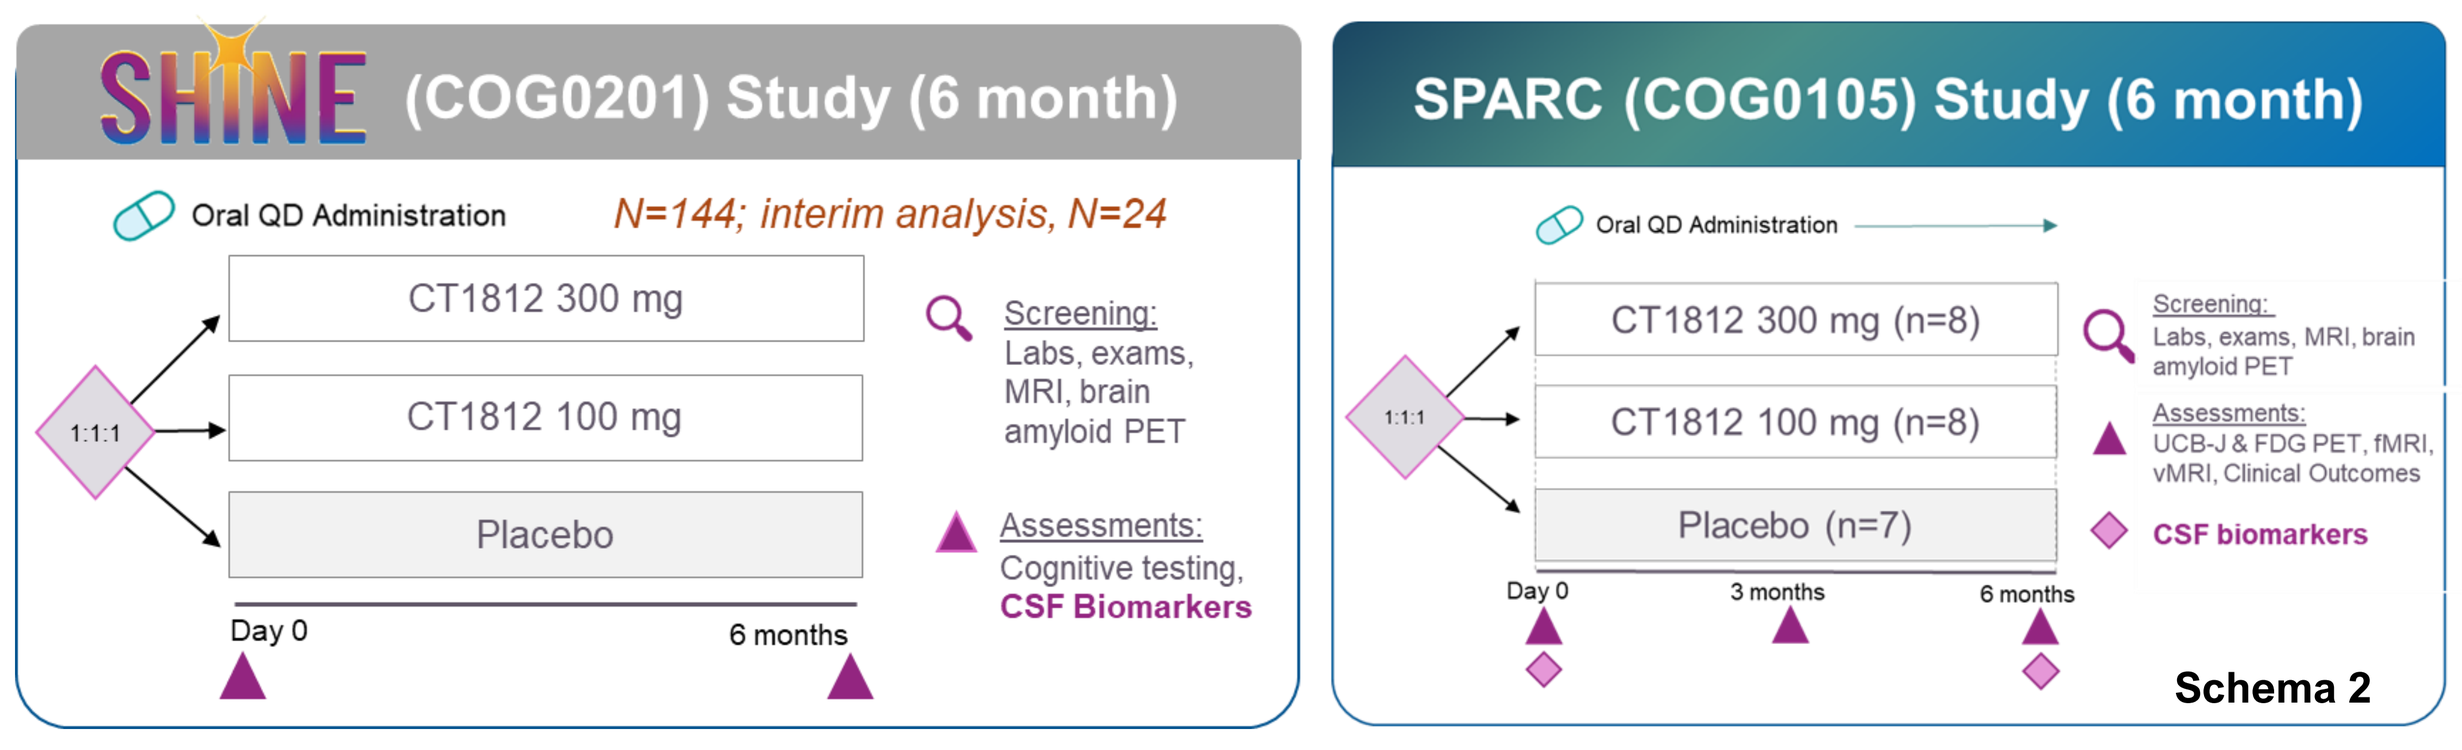
SHINE (COG0201) Study (6 month)
Oral QD Administration
N=144; interim analysis, N=24
1:1:1
CT1812 300 mg
CT1812 100 mg
Placebo
Day 0
6 months
Screening: Labs, exams, MRI, brain amyloid PET
Assessments: Cognitive testing, CSF Biomarkers
SPARC (COG0105) Study (6 month)
Oral QD Administration
CT1812 300 mg (n=8)
CT1812 100 mg (n=8)
Placebo (n=7)
Day 0
3 months
6 months
Screening: Labs, exams, MRI, brain amyloid PET
Assessments: UCB-J & FDG PET, fMRI, vMRI, Clinical Outcomes
CSF biomarkers
Schema 2

Objectives: A meta-analysis of CSF proteomes from Alzheimer's disease (AD) patients from the SHINE and SPARC clinical trials was performed to identify pharmacodynamic biomarkers of the sigma-2 receptor (S2R) modulator CT1812.

Methods: Tandem-mass tag mass spectrometry (TMT-MS) proteomics measurements on CSF from patients included in the interim analysis of the SHINE trial, SHINE-A, and in SPARC were previously performed and the findings presented[2,3]. Although encouraging findings, replication in larger well-powered studies are needed. Towards this end, we performed a meta-analysis herein, leveraging the commonalities across trials: a) same treatment duration (6 mo), same patient population (mild to moderate AD), and same methods used to assess CSF proteomes. Combining, for all patients from which we had a baseline and endpoint CSF draw and that were treatment-compliant, the proteomic data from the two studies, increased the sample size from N=17-18 to N=35, resulting in an increased power for statistical analyses to identify pharmacodynamic biomarkers of CT1812 reaching statistical significance across clinical trial cohorts (**Schema 3**).

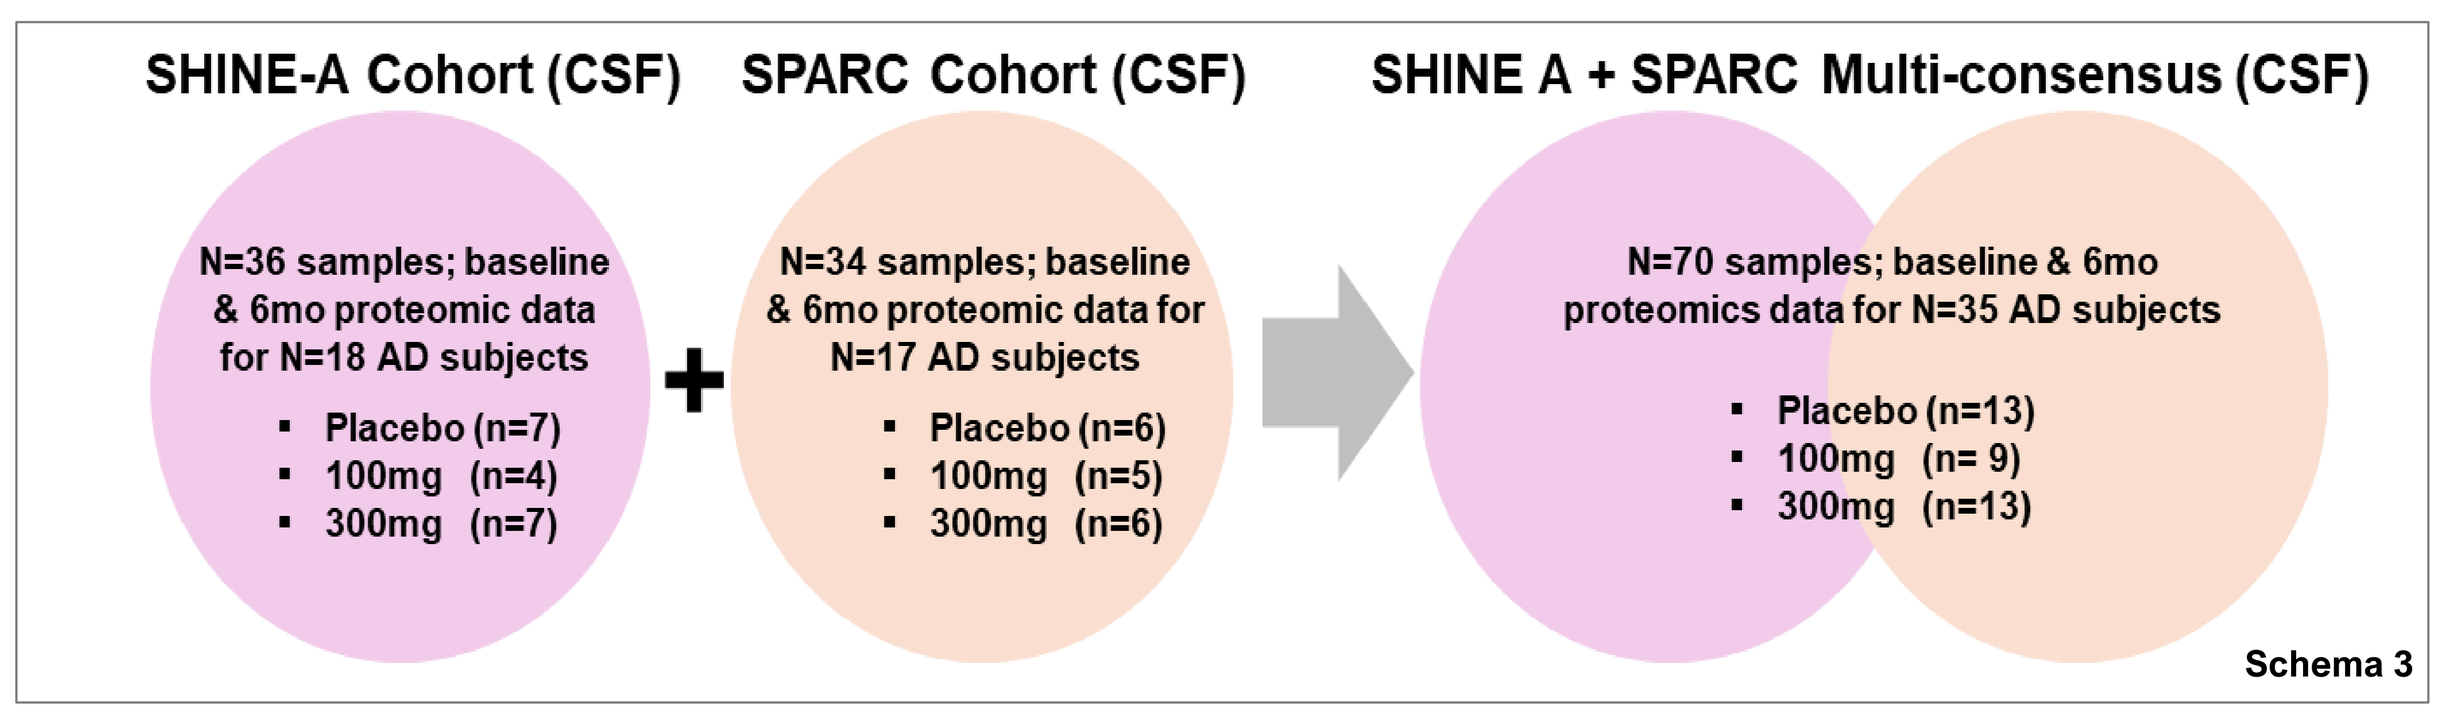
SHINE-A Cohort (CSF)
SPARC Cohort (CSF)
SHINE A + SPARC Multi-consensus (CSF)
N=36 samples; baseline & 6mo proteomic data for N=18 AD subjects
Placebo (n=7)
100mg (n=4)
300mg (n=7)
N=34 samples; baseline & 6mo proteomic data for N=17 AD subjects
Placebo (n=6)
100mg (n=5)
300mg (n=6)
N=70 samples; baseline & 6mo proteomics data for N=35 AD subjects
Placebo (n=13)
100mg (n= 9)
300mg (n=13)
Schema 3

Following preprocessing to remove batch effects, treatment effects of CT1812 (CT1812 vs placebo; p≤0.05) were assessed via differential expression comparative analyses and pathway analyses, using MetaCore and STRING, brain mapping[4], and weighted gene co-expression network analysis (WGCNA) to identify treatment related networks, or modules **(Schema 4)**.

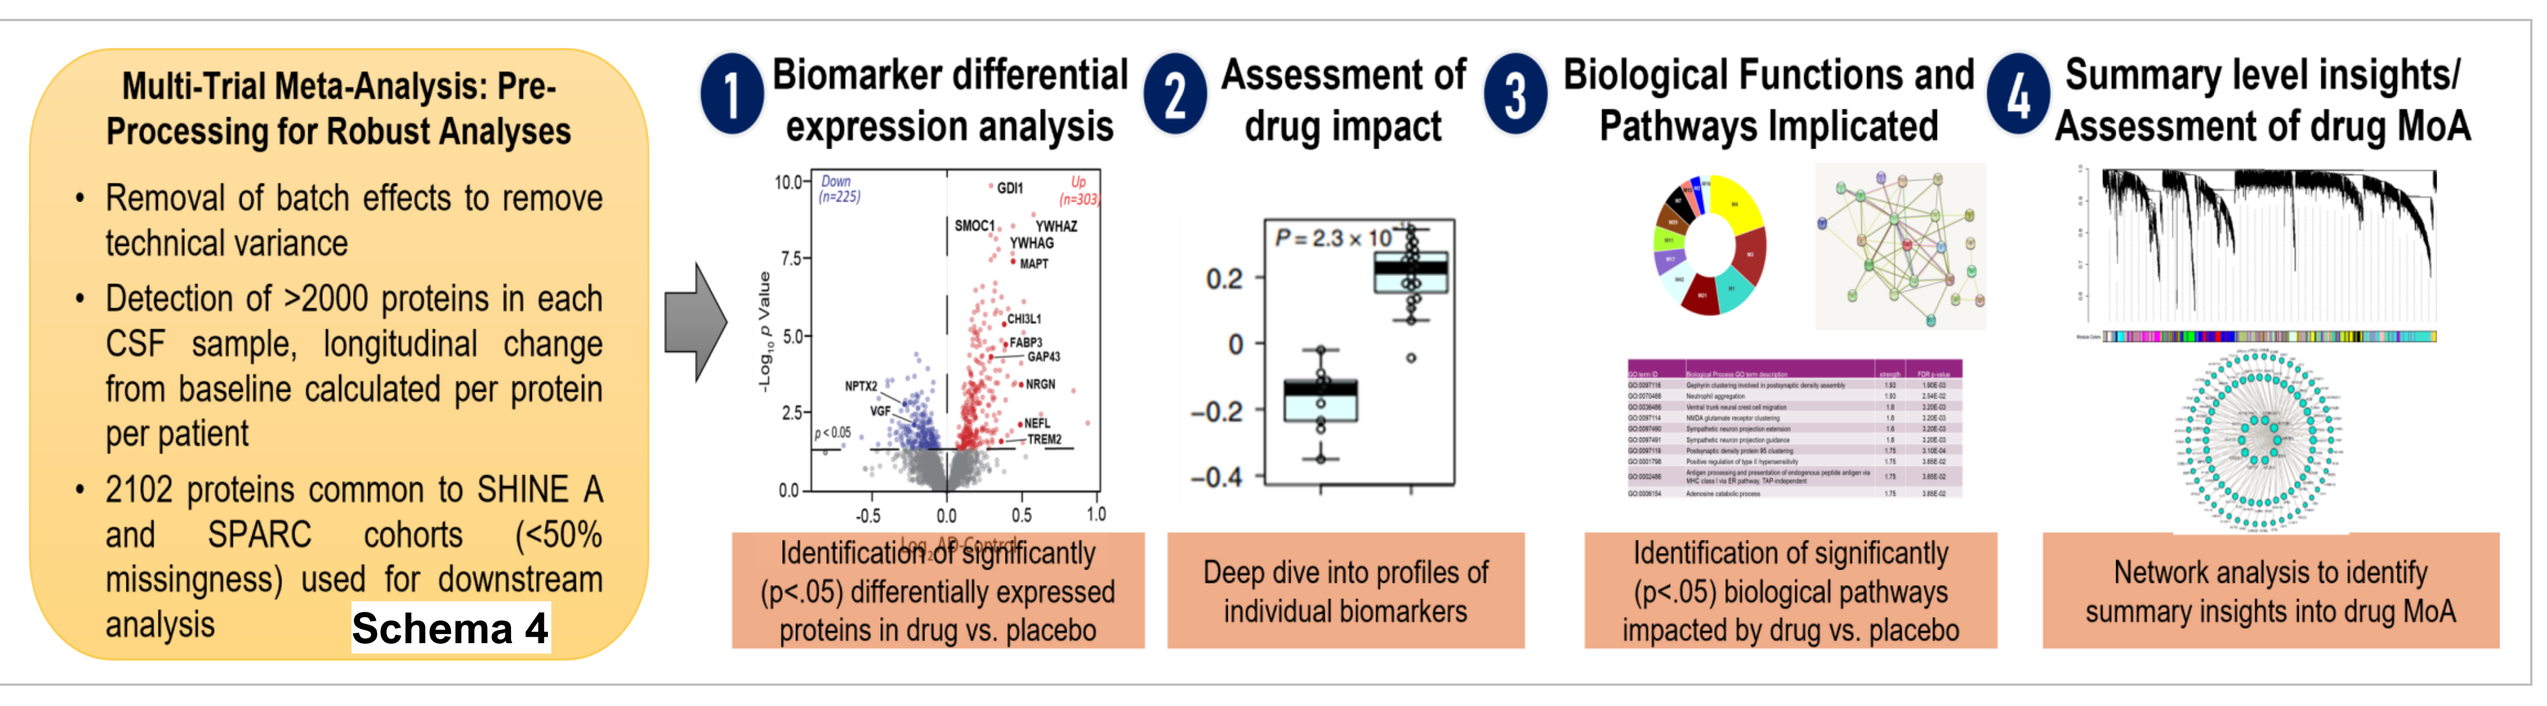
Multi-Trial Meta-Analysis: Pre-Processing for Robust Analyses
Removal of batch effects to remove technical variance
Detection of >2000 proteins in each CSF sample, longitudinal change from baseline calculated per protein per patient
2102 proteins common to SHINE A and SPARC cohorts (<50% missingness) used for downstream analysis
1 Biomarker differential expression analysis
2 Assessment of drug impact
3 Biological Functions and Pathways Implicated
4 Summary level insights/ Assessment of drug MoA
Identification of significantly (p<.05) differentially expressed proteins in drug vs. placebo
Deep dive into profiles of individual biomarkers
Identification of significantly (p<.05) biological pathways impacted by drug vs. placebo
Network analysis to identify summary insights into drug MoA
Schema 4

Pharmacodynamic Biomarkers of CT1812 Identified, & Brain Network Mapping Supports Role of CT1812 at Synapses

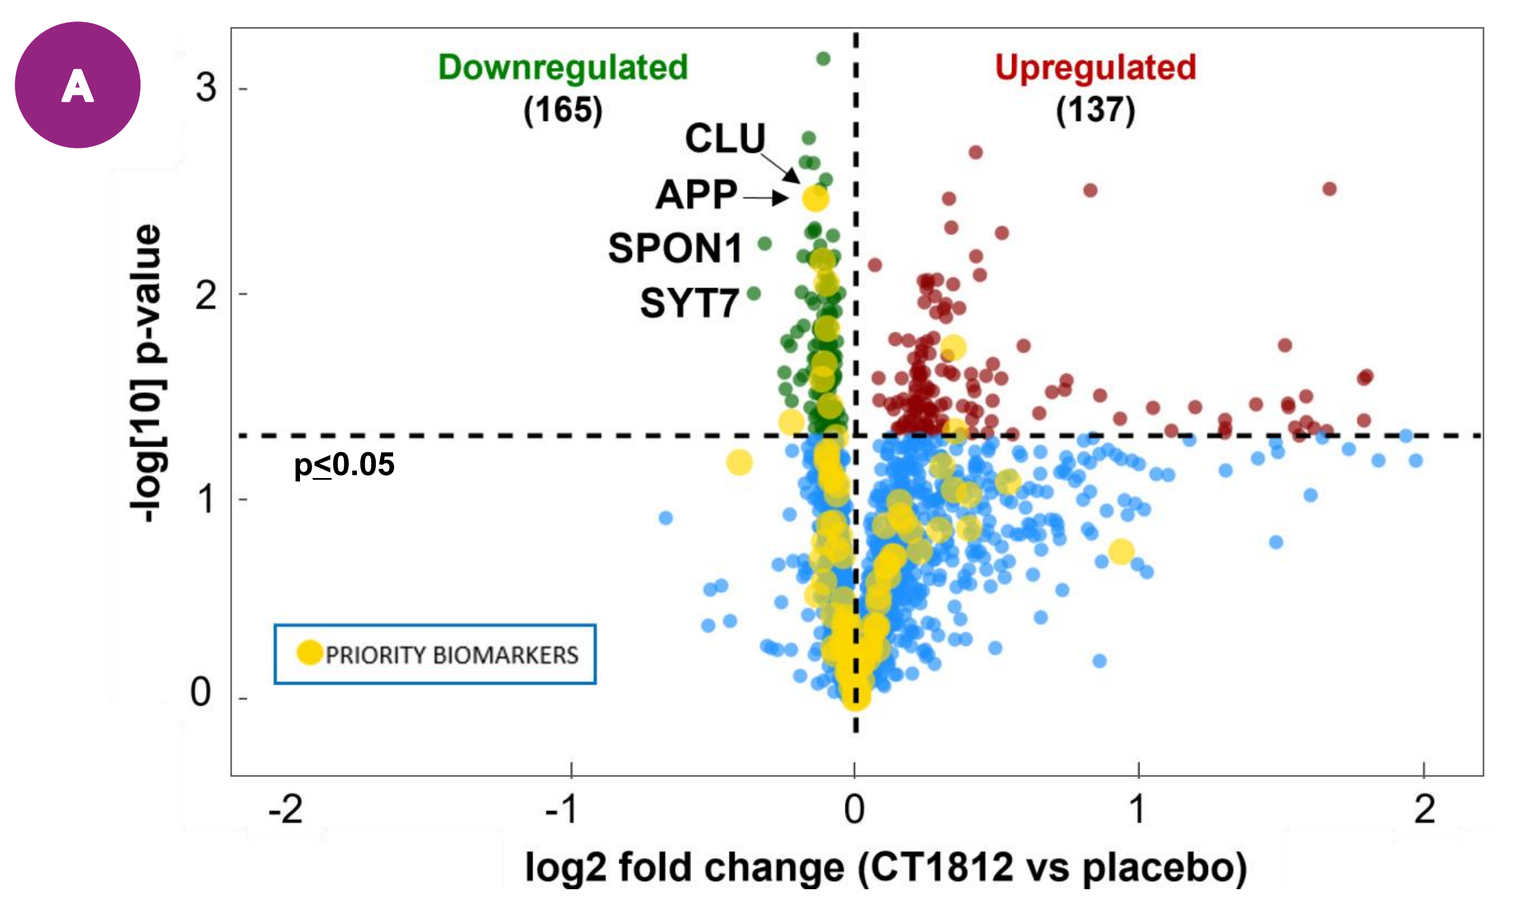
A
Downregulated (165)
Upregulated (137)
CLU
APP
SPON1
SYT7
p≤0.05
PRIORITY BIOMARKERS
-log[10] p-value
log2 fold change (CT1812 vs placebo)

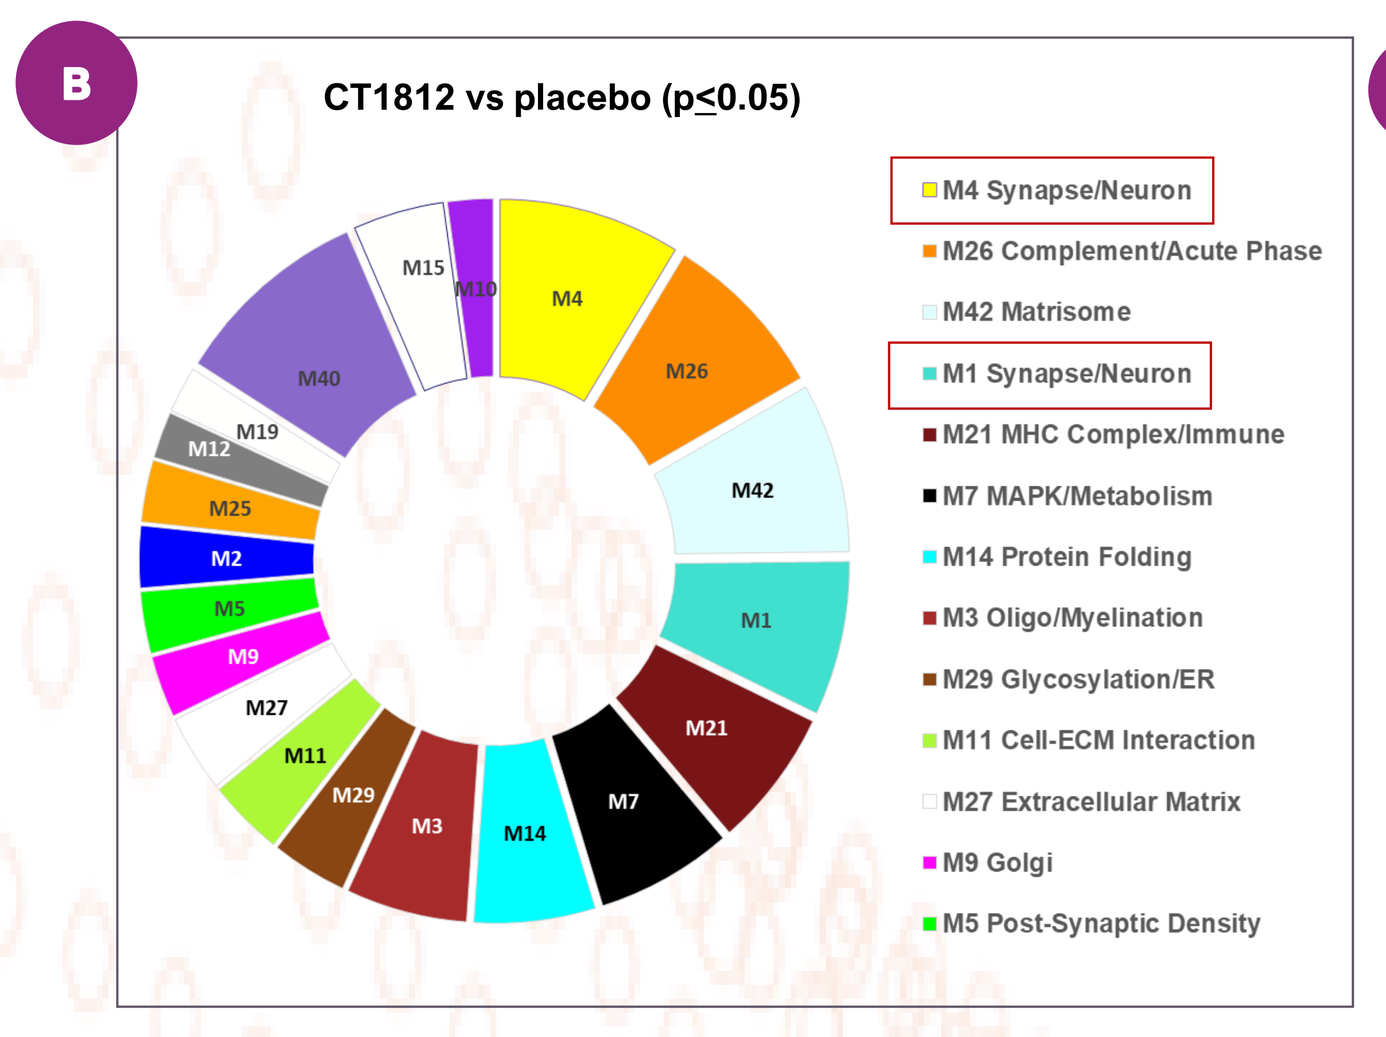
B
CT1812 vs placebo (p≤0.05)
M4 Synapse/Neuron
M26 Complement/Acute Phase
M42 Matrisome
M1 Synapse/Neuron
M21 MHC Complex/Immune
M7 MAPK/Metabolism
M14 Protein Folding
M3 Oligo/Myelination
M29 Glycosylation/ER
M11 Cell-ECM Interaction
M27 Extracellular Matrix
M9 Golgi
M5 Post-Synaptic Density

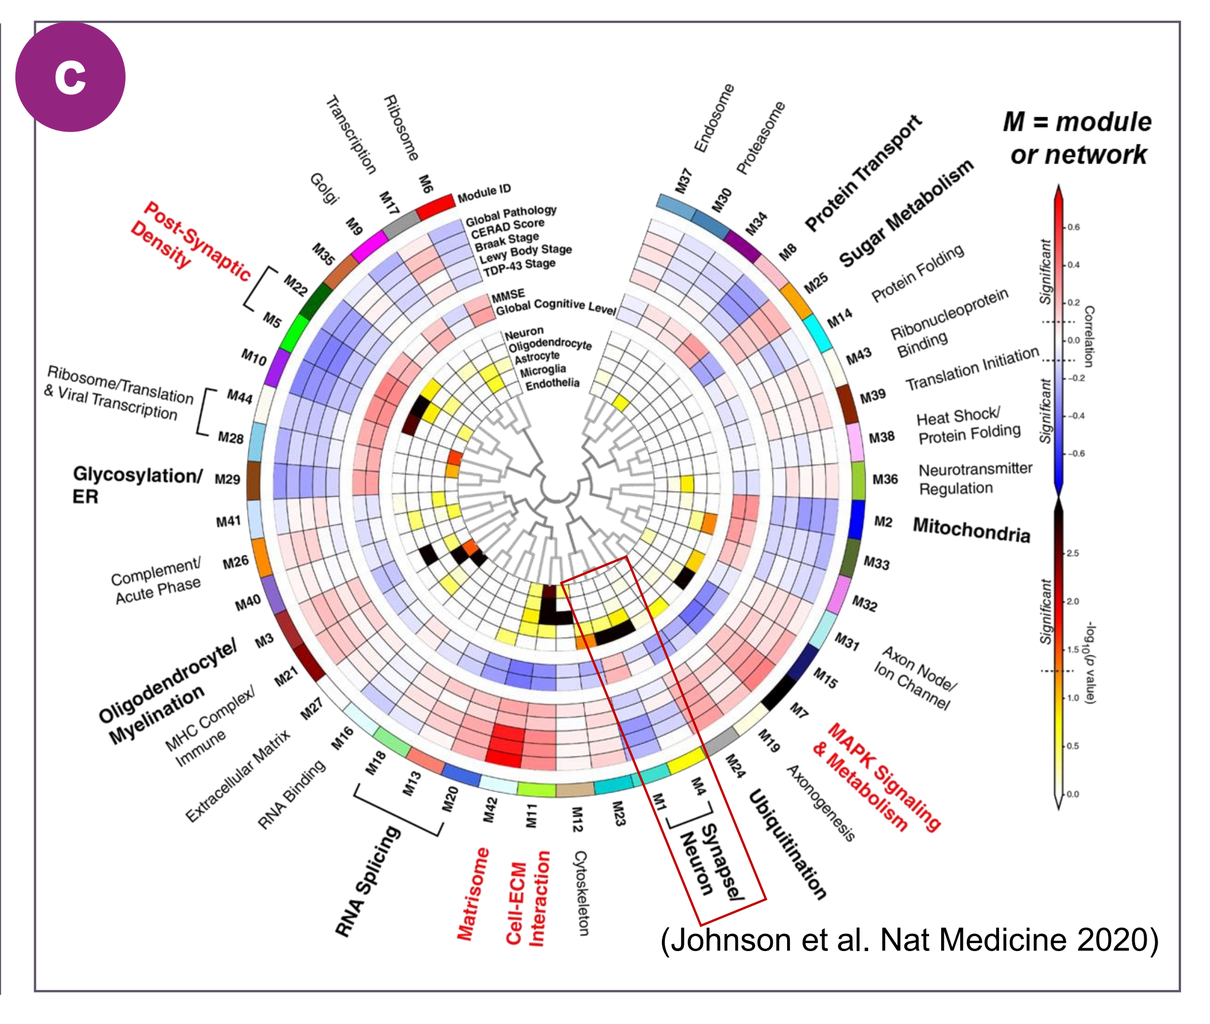
C
M = module or network
Post-Synaptic Density
Synapse/ Neuron
MAPK Signaling & Metabolism
Mitochondria
RNA Splicing
Matrisome
Cell-ECM Interaction
(Johnson et al. Nat Medicine 2020)

Fig 1. A) Volcano plot illustrating 302 proteins were significantly altered (p≤0.05) in CT1812 vs placebo CSF from Alzheimer's disease patients. **B)** Differentially expressed proteins (CT1812 vs placebo; p≤0.05) were mapped to previously generated AD brain co-expression network modules[2] **(C)** built from 516 brain samples with healthy individuals, asymptomatic and symptomatic AD patients to understand how CSF biomarkers altered by CT1812 might impact brain networks disrupted in AD. Top modules represented are indicated **(B)**, with Synapse module M4 the top module identified.

REFERENCES

1. NJ Izzo, CM Yuede, KM LaBarbera, CS Limegrover, C Rehak…ME Hamby, C Williams, K Sadlek, HM Edwards, CS Davis, M Grundman, LS Schneider, ST DeKosky, D Chelsky, I Pike, C Henstridge, K Blennow, H Zetterberg, H Levine, TL Spires-Jones, JR Cirrito, SM Catalano. *Preclinical and clinical biomarker studies of CT1812: A novel approach to Alzheimer's disease modification.* Alz & Dementia 2021; 17: 1365– 1382..
2. Pandey K, Waybright L, Duong DM, Malagise E, Blennow K, Zetterberg H, Mecca AP, van Dyck C, Caggiano AO, Seyfried NT, Hamby ME. *CSF proteomics analysis to investigate the pharmacodynamic response of the S2R modulator CT1812 in Alzheimer's disease patients from the SPARC clinical trial.* AAIC; Alzheimer's & Dementia Volume 18, Issue S6, 18: e068166.
3. N Seyfried, L Waybright, D Duong, E. Malagise, K Pandey, C Williams, E Dammer, L Ping, K Blennow, H Zetterberg, J Lah, A Levey, L Ricciardi, A Caggiano, Hamby ME. *Proteomic Analysis of CSF in a Phase 2 Clinical Trial in Alzheimer's Patients To Identify Pharmacodynamic Biomarkers Of The S2R Modulator CT1812.* AD/PD Advances in Science and Therapy; March 2022, Barcelona Spain.
4. Johnson et. al. *Large-scale deep multi-layer analysis of Alzheimer's disease brain reveals strong proteomic disease-related changes not observed at the RNA level.* Nat Neurosci. 2022.
5. Known AD Biomarkers were sourced from 1) Higginbotham, L et al., *Integrated proteomics reveals brain-based cerebrospinal fluid biomarkers in asymptomatic and symptomatic Alzheimer's disease.* Sci.Adv. (2020); 2) https://www.alzforum.org/alzbiomarker/ad-vs-ctrl

Pathway Analysis of Differentially Abundant Proteins Implicates CT1812 in Regulating Synapses, Neuroinflammation, & Amyloid Biology

A *Gene Ontology (GO) analysis further highlights role of CT1812 in regulating synapse biology*

Top Biological Processes (GO Terms); CT1812 vs placebo; p≤0.05

GO term ID	Biological Process GO term description	strength	FDR p-value
GO:0097116	Gephyrin clustering involved in postsynaptic density assembly	1.93	1.90E-03
GO:0070488	Neutrophil aggregation	1.93	2.54E-02
GO:0036486	Ventral trunk neural crest cell migration	1.8	3.20E-03
GO:0097114	NMDA glutamate receptor clustering	1.8	3.20E-03
GO:0097490	Sympathetic neuron projection extension	1.8	3.20E-03
GO:0097491	Sympathetic neuron projection guidance	1.8	3.20E-03
GO:0097119	Postsynaptic density protein 95 clustering	1.75	3.10E-04
GO:0001798	Positive regulation of type II hypersensitivity	1.75	3.85E-02
GO:0002486	Antigen processing and presentation of endogenous peptide antigen via MHC class I via ER pathway, TAP-independent	1.75	3.85E-02
GO:0006154	Adenosine catabolic process	1.75	3.85E-02

B *Metacore Pathway Analysis indicates role of CT1812 in regulating amyloid biology and inflammation*

	Top Pathways (CT1812 vs placebo; p≤0.05)	FDR p-value
** 1	Gamma-secretase proteolytic targets	2.87E-05
2	Immune response_Lectin induced complement pathway	5.16E-04
3	Immune response_Classical complement pathway	5.16E-04
4	Immune response_Alternative complement pathway	5.16E-04
** 5	Gamma-secretase regulation of neuronal cell development and function	6.79E-04
6	Alternative complement cascade disruption in age-related macular degeneration	3.22E-03
* 7	O-Glycan biosynthesis	8.46E-03
8	N-Glycan biosynthesis p2	1.08E-02
9	Neurophysiological process_Dynein-dynactin motor complex in axonal transport in neurons	2.85E-02
** 10	Aberrant lipid trafficking and metabolism in age-related macular degeneration pathogenesis	4.21E-02

Fig 2. A) Gene ontology (GO) analysis and **(B)** Metacore pathway mapping of differentially abundant proteins (CT1812 vs placebo; p≤0.05) was performed. * indicates pathways previously identified as impacted by CT1812 vs placebo in the SPARC or SHINE-A independent cohort analyses. Displayed (**A**-**B**) are the top-most statistically significant pathways altered by CT1812.

Priority AD Biomarkers Altered by CT1812, and Clear Separation Of Patients by Treatment (CT1812 vs Placebo)

A *Priority Biomarkers Impacted by CT1812*

CT1812 vs Placebo	Direction of change [1]	p val. (unadjusted[2])	REF. AD-CT[3]
CLU\|P10909-4	↓	0.003	↑
APP\|P05067	↓	0.003	↓
SPON1\|Q9HCB6	↓	0.007	↑
COL6A1\|P12109	↓	0.009	↓
DDAH1\|O94760	↓	0.015	↑
YWHAB\|P31946	↑	0.018	↑
APOE\|P02649	↓	0.026	↓
OLFML3\|Q9NRN5	↓	0.022	↑
PTPRZ1\|P23471	↓	0.036	↑
SPP1\|P10451–5	↓	0.043	↑
GLOD4\|Q9HC38–2	↑	0.048	↑

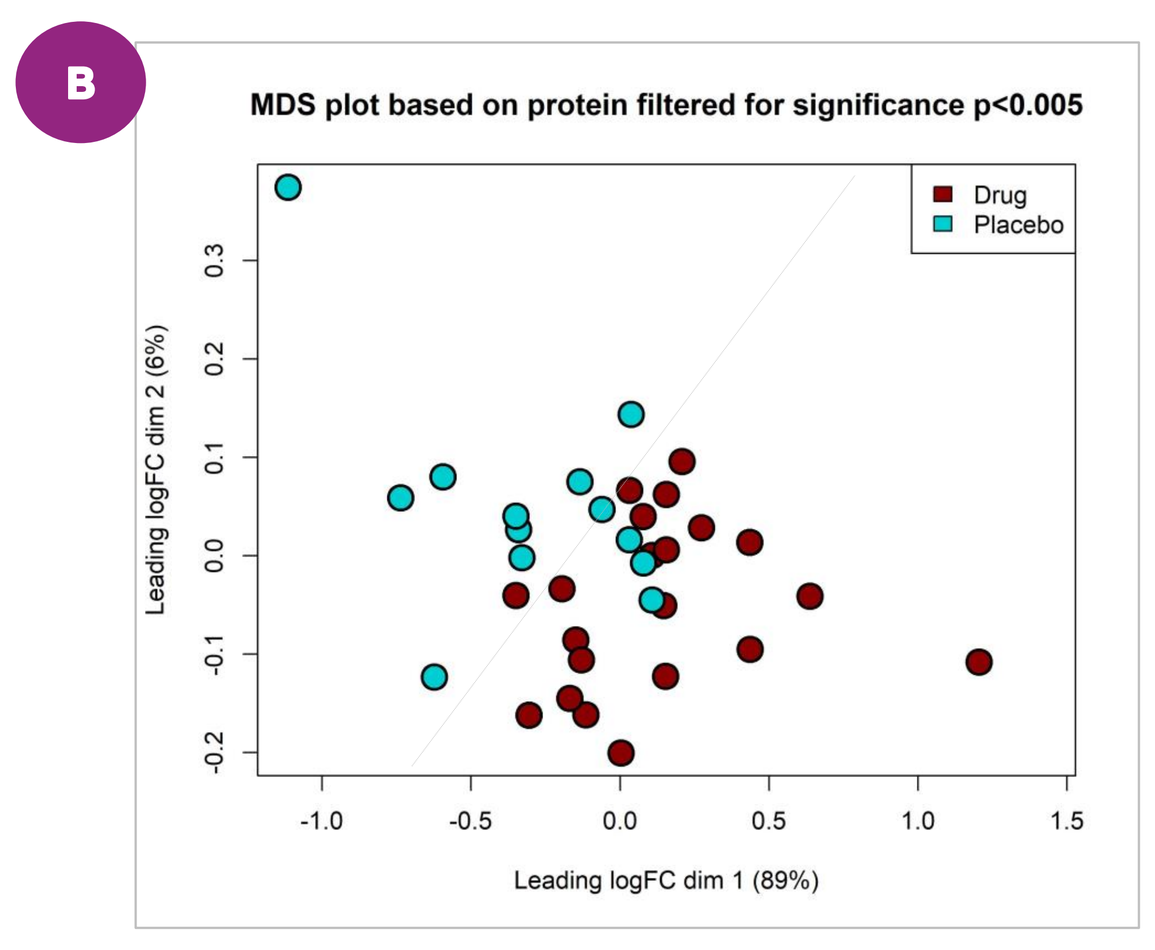
B
MDS plot based on protein filtered for significance p<0.005
Drug
Placebo
Leading logFC dim 2 (6%)
Leading logFC dim 1 (89%)

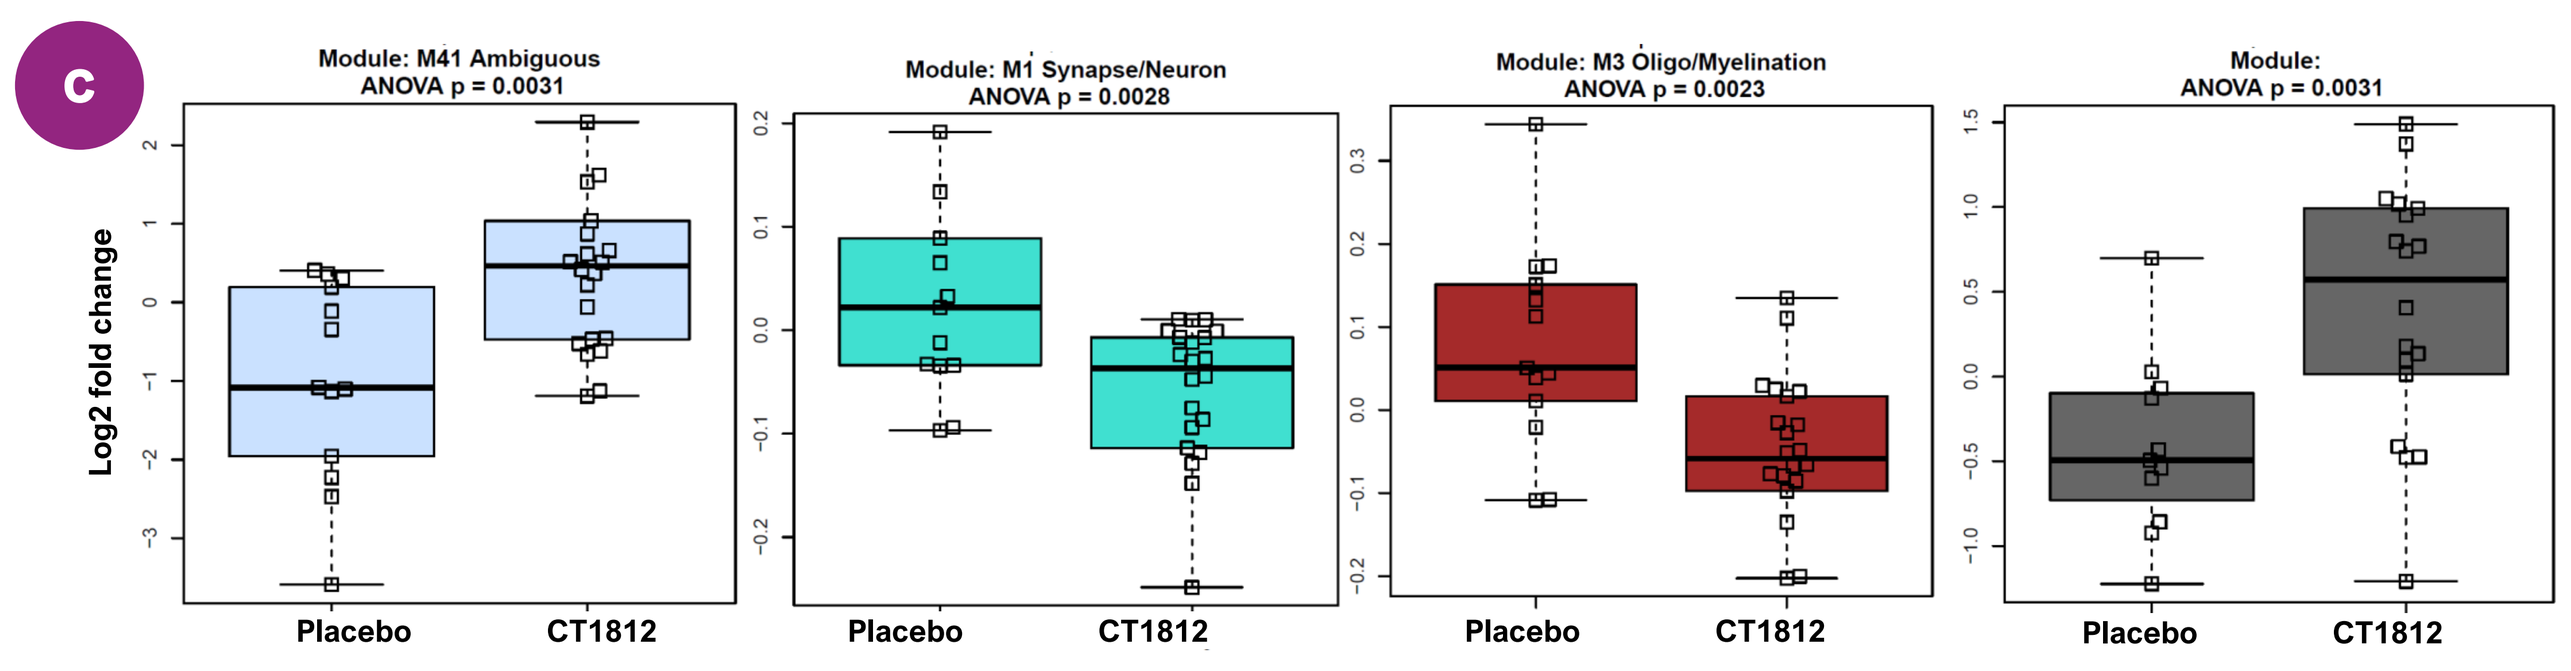
C
Module: M41 Ambiguous ANOVA p = 0.0031
Module: M1 Synapse/Neuron ANOVA p = 0.0028
Module: M3 Oligo/Myelination ANOVA p = 0.0023
Module: ANOVA p = 0.0031
Log2 fold change
Placebo
CT1812

Fig 3. A) 11 AD priority biomarkers[5] were found to be significantly regulated by CT1812 vs placebo. **B)** Multidimensional scaling (MDS) plot demonstrates the level of similarity of individual patient CSF biomarker data using a p<0.005 criterion. **C)** Example biomarkers of most highly significantly altered proteins illustrate robust treatment effects

Comparative Analyses Illuminate Robust Candidate Pharmacodynamic Biomarkers that Replicate Across Independent Cohorts / Analyses

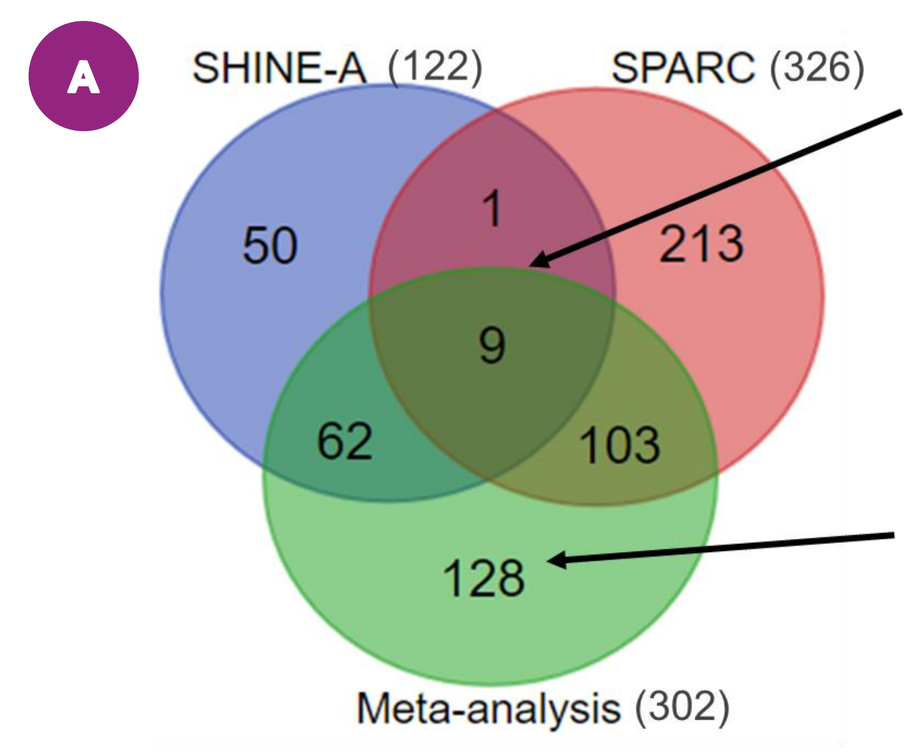
A
SHINE-A (122)
SPARC (326)
50
1
213
9
62
103
128
Meta-analysis (302)

- Replication of biomarker changes by CT1812 vs placebo across two independent cohorts
 - but not in meta-analysis; (**1**) NPC1 – significance observed in both trials, but change in opposing direction
 - and significance maintained in meta-analysis (**9**)

- 128 biomarkers that may require increased power to detect significance
 - 19 of these demonstrated a trend p≤0.10 in both trials (see table below)

B

	Meta-analysis		SHINE-A		SPARC	
Protein	p-value	Log2 FC	p-value	Log2 FC	p-value	Log2 FC
Protein A	1.00E-02	-0.36	6.56E-02	-0.40	5.57E-02	-0.34
Protein B	3.64E-02	-0.16	5.43E-02	-0.14	9.73E-02	-0.20
Protein C	2.38E-02	-0.15	8.30E-02	-0.14	7.38E-02	-0.19
Protein D	1.54E-02	-0.13	5.66E-02	-0.16	5.62E-02	-0.12
Protein E	2.61E-02	-0.13	6.51E-02	-0.16	5.91E-02	-0.13
Protein F	2.64E-02	-0.12	8.50E-02	-0.14	7.47E-02	-0.13
Protein G	1.49E-02	-0.10	9.33E-02	-0.11	9.06E-02	-0.11
Protein H	2.06E-02	-0.10	7.90E-02	-0.12	9.37E-02	-0.08
Protein I	2.29E-02	-0.09	7.63E-02	-0.08	6.66E-02	-0.12
Protein J	1.24E-02	-0.09	6.85E-02	-0.08	5.55E-02	-0.12
Protein K	1.79E-02	-0.09	6.38E-02	-0.11	9.28E-02	-0.09
Protein L	1.99E-02	-0.08	6.82E-02	-0.09	7.61E-02	-0.09
Protein M	3.60E-02	-0.08	9.56E-02	-0.08	9.82E-02	-0.10
Protein N	1.23E-02	-0.07	5.03E-02	-0.08	6.37E-02	-0.09
Protein O	2.84E-02	0.21	8.97E-02	0.19	5.40E-02	0.30
Protein P	1.72E-02	0.25	8.69E-02	0.23	9.57E-02	0.26
Protein Q	1.19E-02	0.31	6.54E-02	0.22	5.51E-02	0.40
Protein R	1.13E-02	0.32	6.87E-02	0.21	5.48E-02	0.42
Protein S	2.22E-02	0.48	9.47E-02	0.47	9.31E-02	0.50

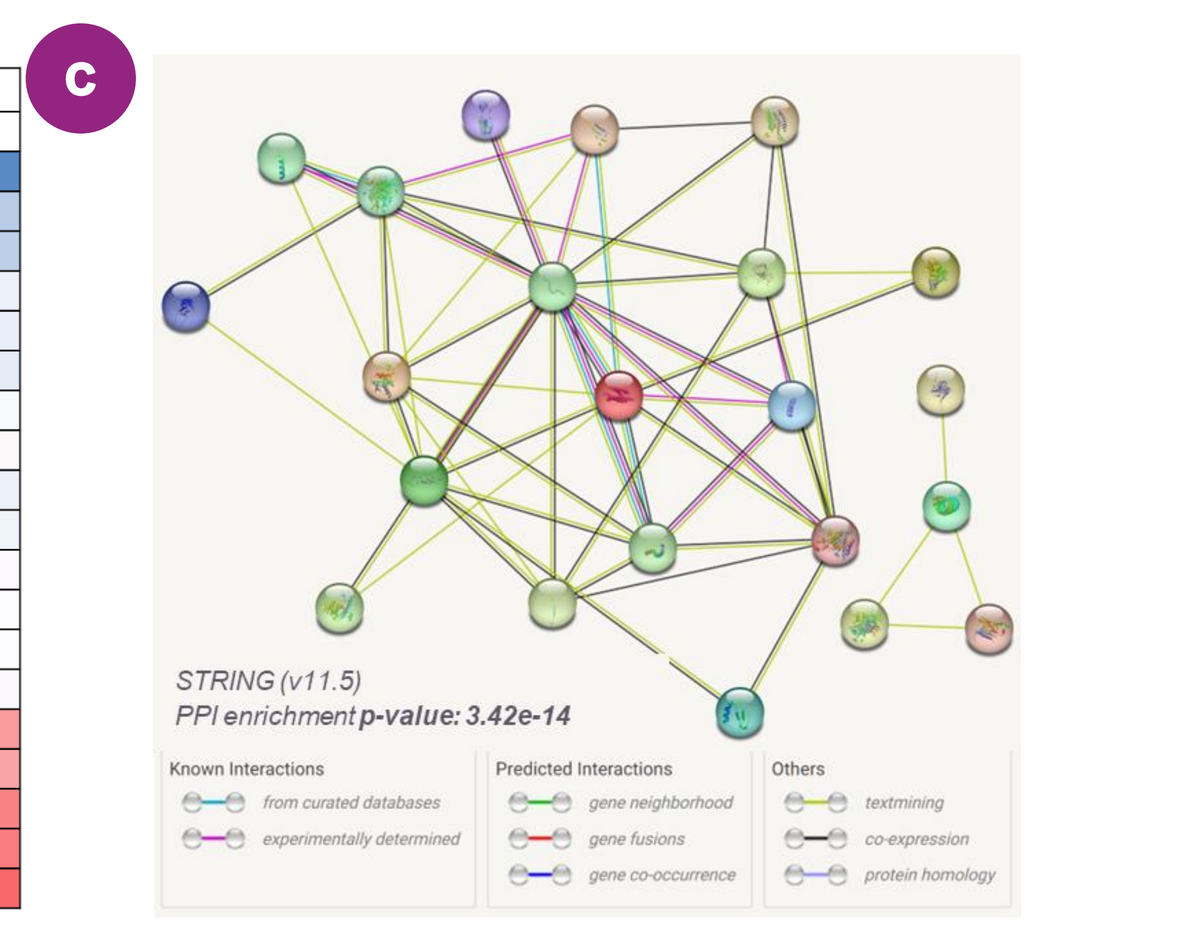
C
STRING (v11.5)
PPI enrichment p-value: 3.42e-14
Known Interactions
from curated databases
experimentally determined
Predicted Interactions
gene neighborhood
gene fusions
gene co-occurrence
Others
textmining
co-expression
protein homology

D

GO term	Biological Process (Replicated Biomarkers; CT1812 vs placebo)	strength	FDR p-value
GO:1902003	Regulation of amyloid-beta formation	2.14	6.00E-06
GO:1902430	Negative regulation of amyloid-beta formation	2.41	1.55E-05
GO:1902993	Positive regulation of amyloid precursor protein catabolic process	2.17	6.17E-05
GO:1905908	Positive regulation of amyloid fibril formation	2.82	9.19E-05
GO:0050808	Synapse organization	1.28	0.00089
GO:1900272	Negative regulation of long-term synaptic potentiation	2.35	0.00089
GO:1902947	Regulation of tau-protein kinase activity	2.31	0.00089
GO:1902950	Regulation of dendritic spine maintenance	2.35	0.00089
GO:0048638	Regulation of developmental growth	1.2	0.0015
GO:1900221	Regulation of amyloid-beta clearance	2.2	0.0015

Fig 4. A) A comparative analysis assessing the overlap of biomarkers identified from independent cohorts and analyses was performed. Biomarker replication (statistical significance and directionality) across two independent cohorts was observed (9 biomarkers). 19 biomarkers found to be significant in the meta-analysis just missed significance (p<0.10) in both cohorts when analyzed alone; see table (**B**). **C-D)** Pathway analysis was performed on the most robust (9 + 19) biomarkers, which showed a significant STRING network interaction **(C)** and revealed that the most robust biomarkers are linked to synaptic and Aβ biology.

Network Analysis Identifies Treatment-Associated Networks, Hub Proteins Illuminating Mechanistic Insights of CT1812 in AD patients

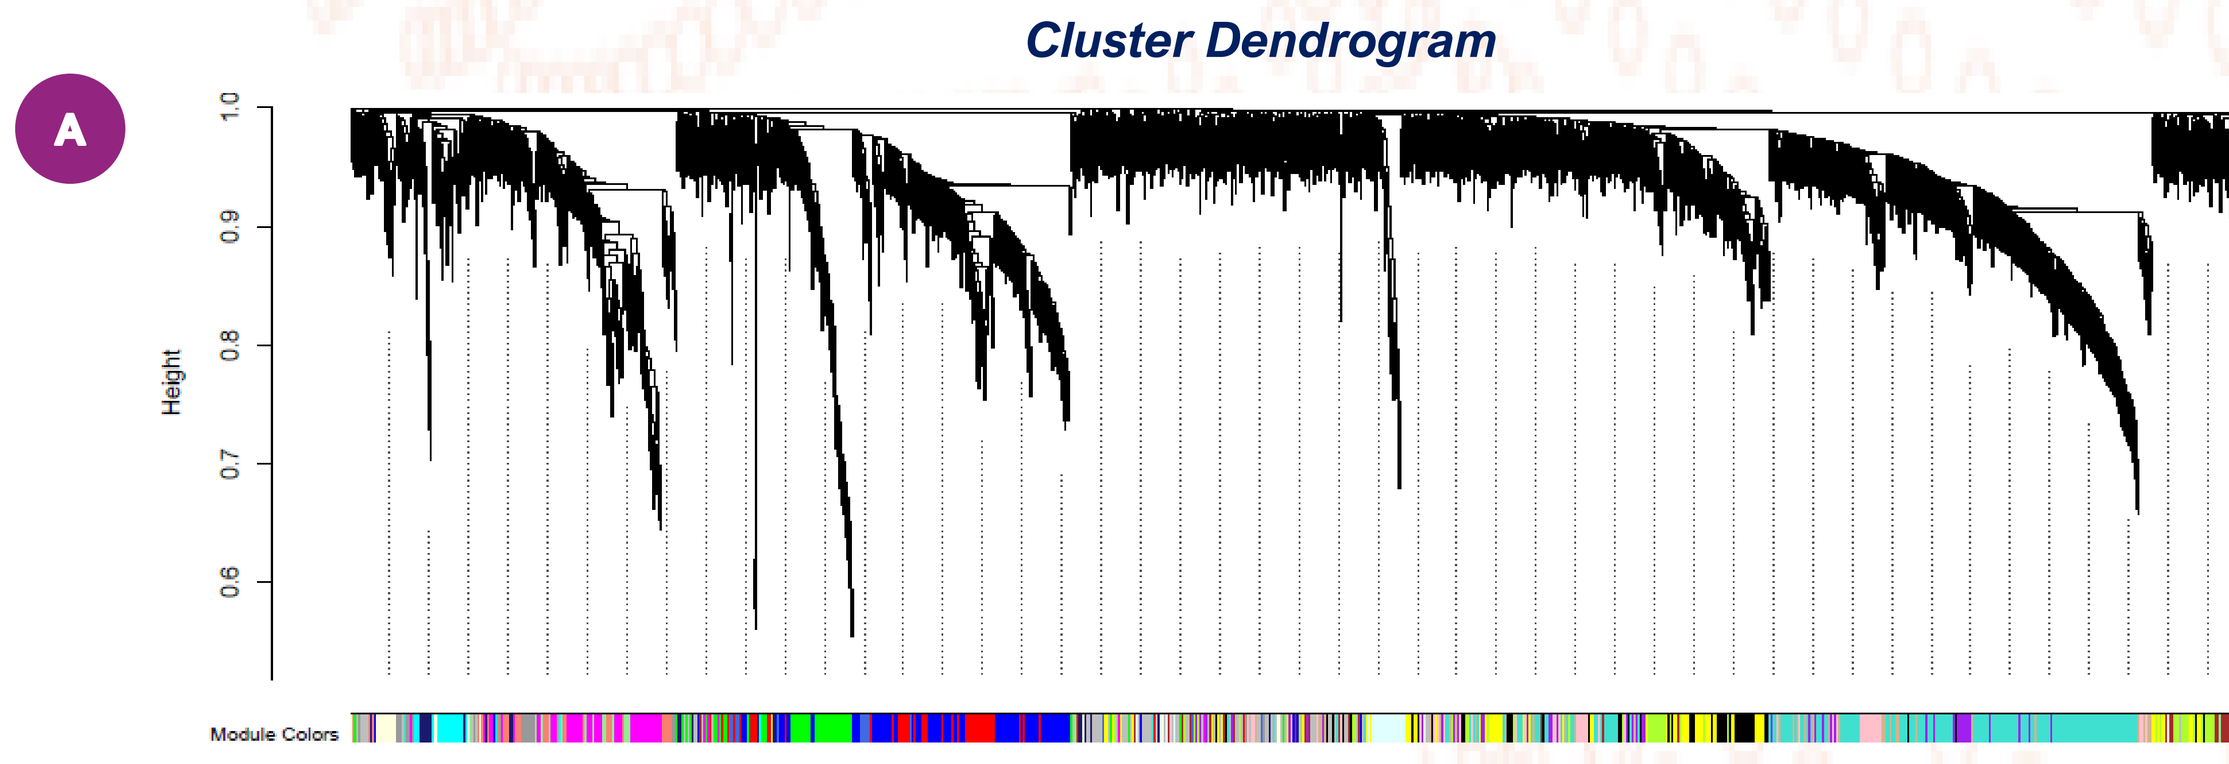
Cluster Dendrogram
A
Height
Module Colors

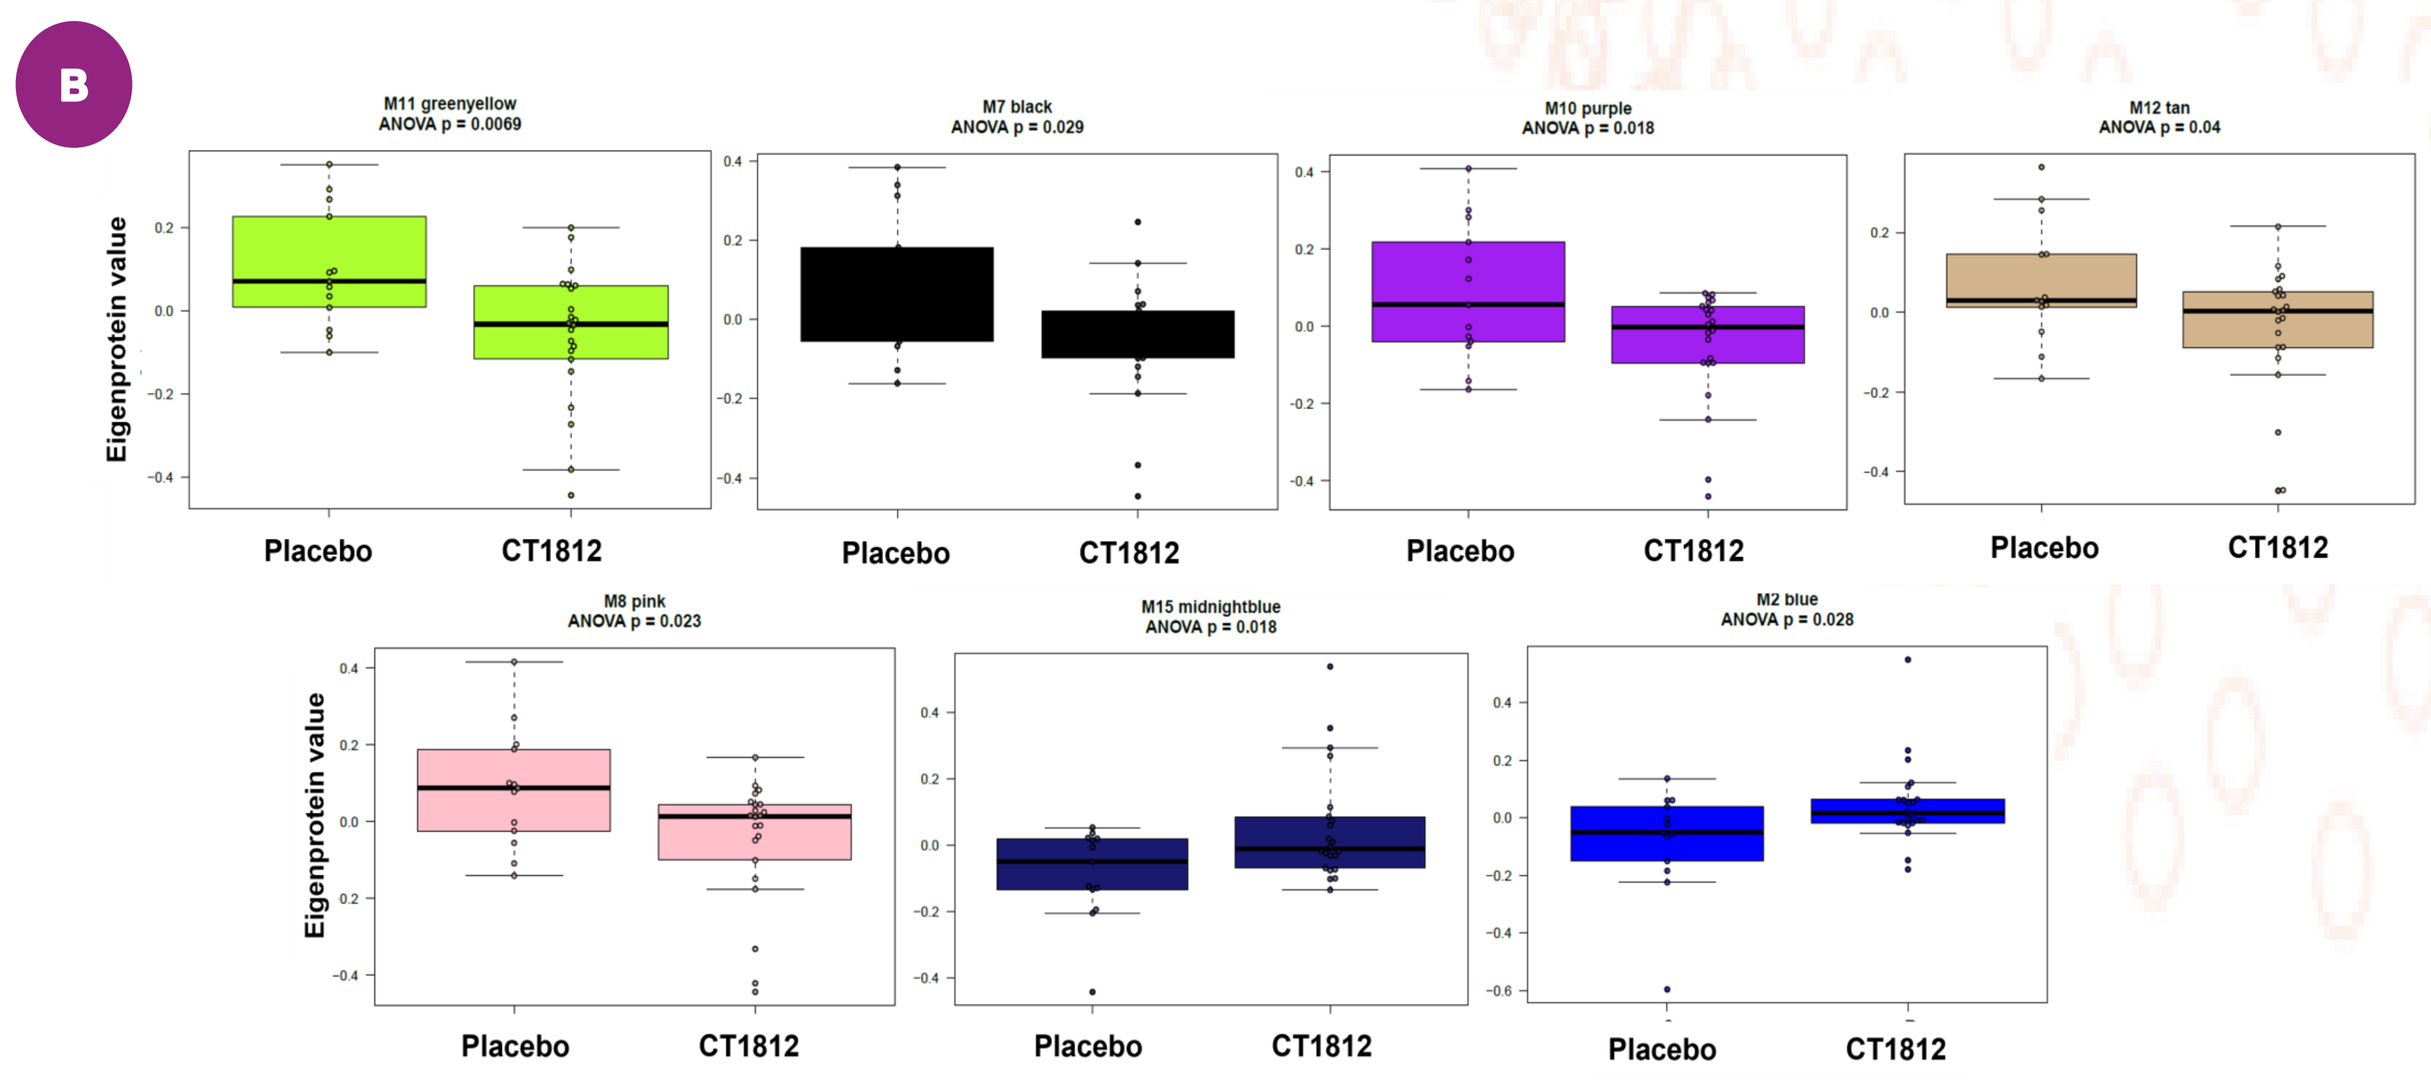
B
M11 greenyellow ANOVA p = 0.0069
M7 black ANOVA p = 0.029
M10 purple ANOVA p = 0.018
M12 tan ANOVA p = 0.04
M8 pink ANOVA p = 0.023
M15 midnightblue ANOVA p = 0.018
M2 blue ANOVA p = 0.028
Eigenprotein value
Placebo
CT1812

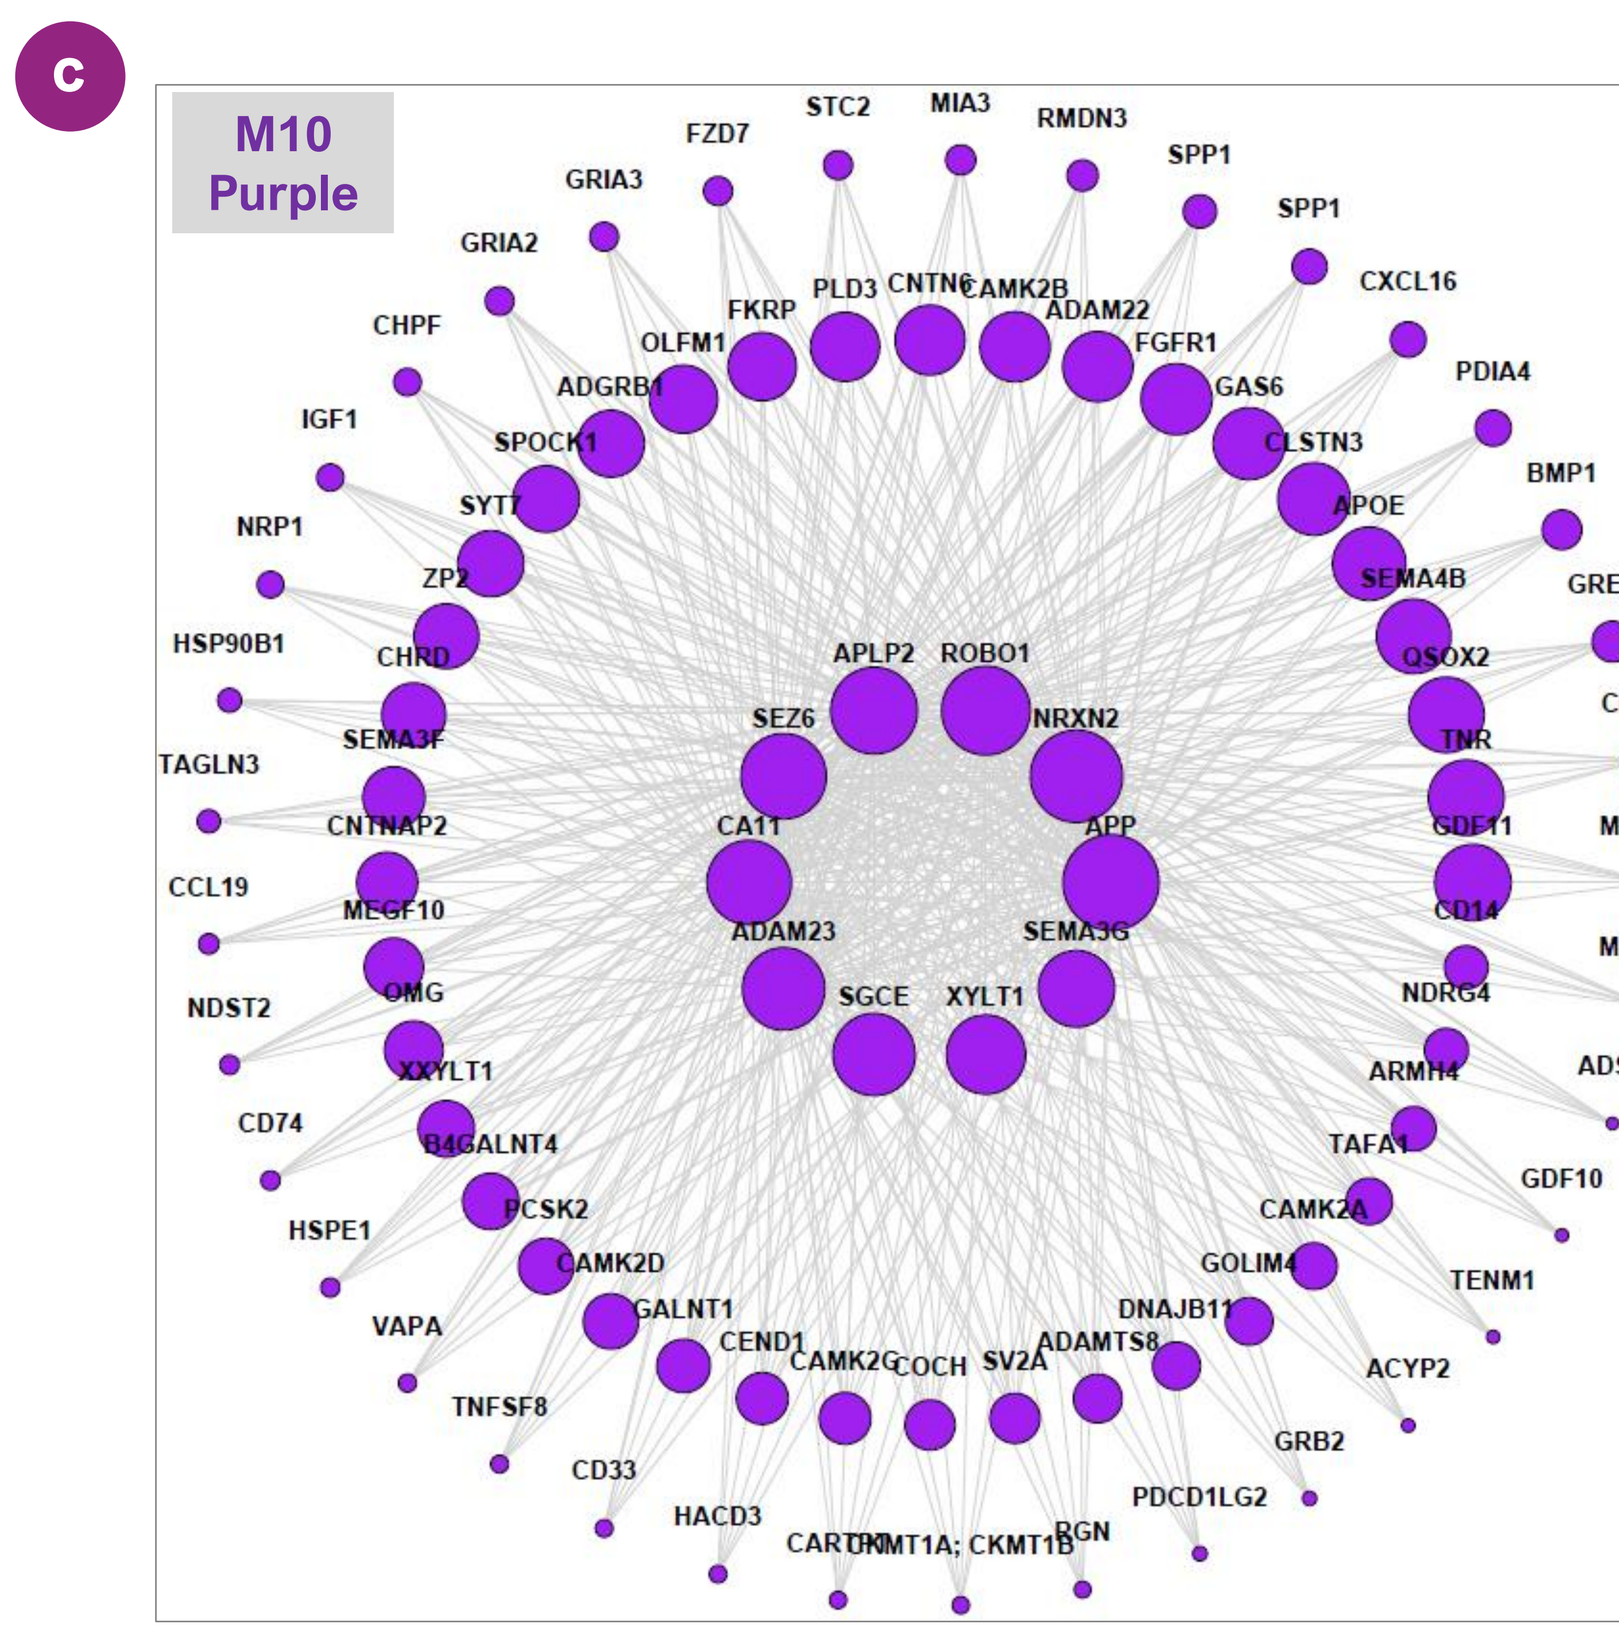
C
M10 Purple

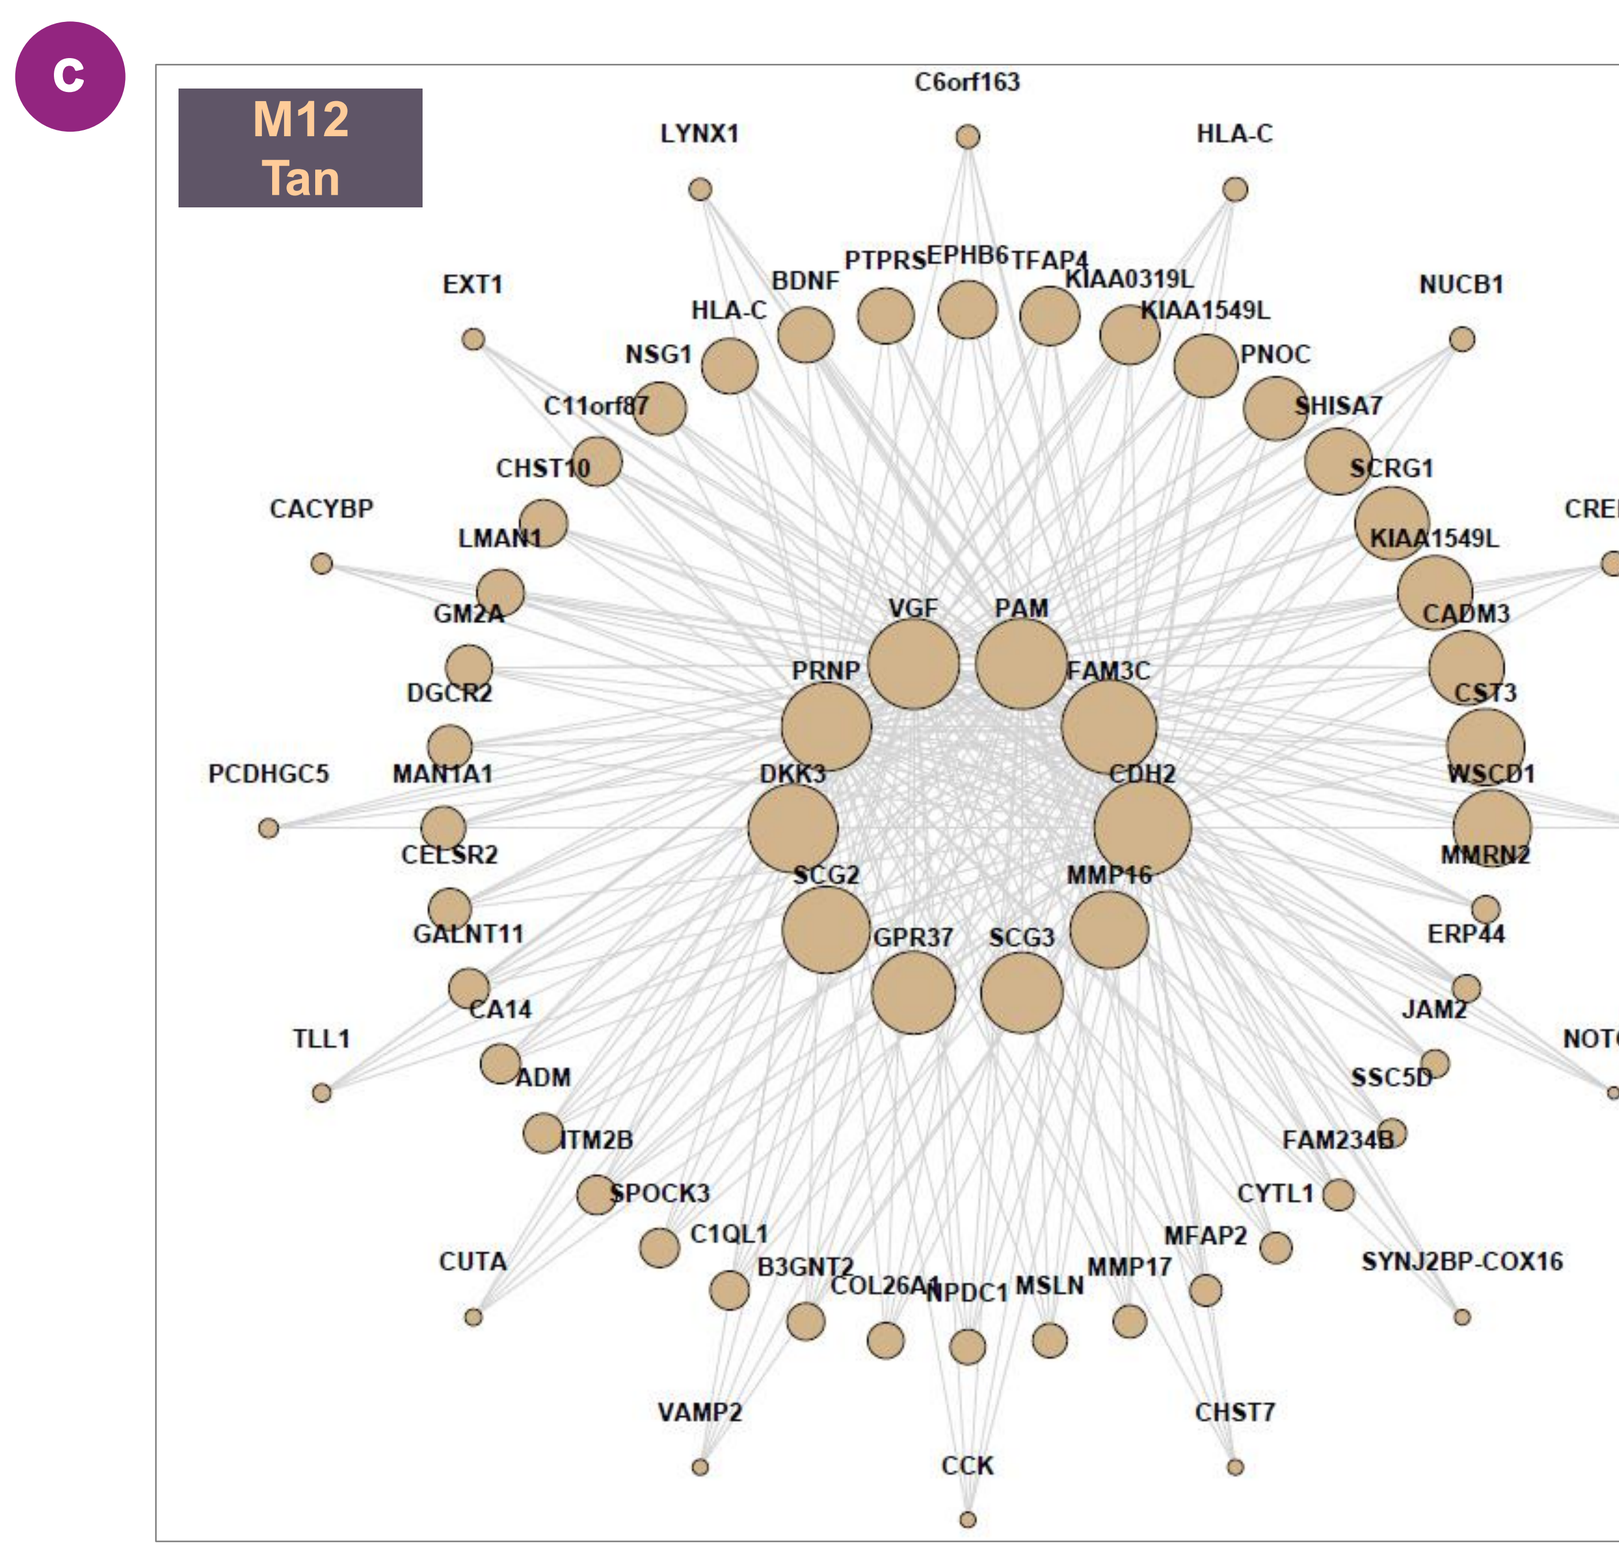
C
M12 Tan

Fig 5. WGCNA was used to identify networks (modules) of proteins that are co-expressed longitudinally across patients. **A)** Hierarchical clustering dendrogram showing clustering of proteins into color modules (20 networks). **B)** Seven out of 20 modules were significantly associated with treatment (CT1812 vs placebo). **C)** Hub proteins (inner circle) with greatest association (kME), along with other proteins in module in the treatment-associated purple and tan modules.

CONCLUSIONS

- This meta-analysis allowed for the identification of robust pharmacodynamic biomarkers of CT1812
 - 302 pharmacodynamic changes due to CT1812 identified
 - 11 AD Priority biomarkers altered by CT1812 vs placebo
 - 9 biomarkers showed replication across trials with similar directionality and degree of change; 19 additional robust biomarkers needing larger sample size to achieve significance discovered
- Brain network mapping and pathway analysis of differentially abundant proteins supports role of CT1812 in synaptic biology, neuroinflammation & amyloid biology
- Comparative analyses illuminate biomarkers that replicate across independent cohorts and analyses, and pathway analysis of these robust biomarkers highlight a prominent role of CT1812 in regulating synaptic and Aβ biology
- Network analysis identifies networks that are associated with CT1812 treatment, enabling further mechanistic understanding of how CT1812 may impact patients with Alzheimer's disease
 - Networks altered include Hub proteins Prion protein (PRNP) known to interact with S2R, and APP, known to be regulated by PRNP

Overall, data provide additional support of a synaptoprotective mechanism of action for CT1812, and support the continued clinical development of CT1812 for Alzheimer's disease

ClinicalTrials.gov: NCT03507790, NCT03493282
Supported by National Institute on Aging: 3R01AG058660, AG057553

Corresponding author: mhamby@cogrx.com