Plasma proteomic analysis of a biomarker-defined subpopulation in the SHINE Ph2 trial to identify molecular correlates to the favorable decrease in the neuroinflammatory marker GFAP with zervimesine in Alzheimer's disease participants

Britney N. Lizama[1], Valentina Di Caro[1], Eunah Cho[1], Jill Caldwell[1], Kiran Pandey[2], Duc Duong[2], Nicholas Seyfried[2], Michael Grundman[3,4], Anthony O. Caggiano[5], Charlotte Teunissen[6], **Mary E. Hamby**[1]

(1) Cognition Therapeutics, Inc., Pittsburgh, PA, USA, (2) Emory University School of Medicine, Atlanta, GA, USA, (3) Global R&D Partners, LLC, La Jolla, CA, USA, (4) University of California, San Diego, CA, USA, (5) Cognition Therapeutics, Inc., Purchase, NY, USA, (6) Neurochemistry Laboratory, Department of Laboratory Medicine, Amsterdam Neuroscience, Vrije Universiteit Amsterdam, Amsterdam UMC, Amsterdam, Netherlands

COGNITION Therapeutics

Poster # 107075

Key takeaway: Robust plasma biomarker changes seen with zervimesine (CT1812) treatment, including a dampening of the neuroinflammation biomarker GFAP, in a pre-specified subgroup with lower levels of AD pathology (low p-tau217)

Background

The sigma-2 receptor (S2R) modulator, zervimesine (CT1812), is an allosteric Aβ oligomer antagonist currently in Phase 2 clinical trials[1] for Alzheimer's disease (AD) and dementia with Lewy bodies[2]. Preclinical and clinical studies have shown that zervimesine displaces Aβ and α-synuclein oligomers from neurons[3], preserving synapses and restoring cognitive performance in a transgenic mouse model of AD[4].

Participants with AD treated with zervimesine exhibited 38% slowing of cognitive decline (ADAS-Cog11) compared to placebo in the mITT cohort of the SHINE trial (NCT03507790, COG0201). However, in the pre-specified below-median p-tau217 subgroup a more robust, 95% slowing of cognitive decline was observed with zervimesine, significant decrease in plasma-GFAP, a neuroinflammation biomarker, compared to placebo (-28.35±11.7 SEM, p=0.019) and trends of reductions for Aβ42, Aβ40 and NfL (p<0.10)[5]. Given favorable outcomes with zervimesine treatment in the below-median p-tau217 subgroup (hereafter referred to as the low p-tau217 group), a plasma proteomic biomarker sub-study assessing correlation of plasma proteins with plasma-GFAP was performed to elucidate neuroinflammatory correlates and S2R-related pharmacodynamic mechanisms of zervimesine.

Trial Design: a Phase 2 Safety and Efficacy Study in Adults with Mild-to-Moderate AD

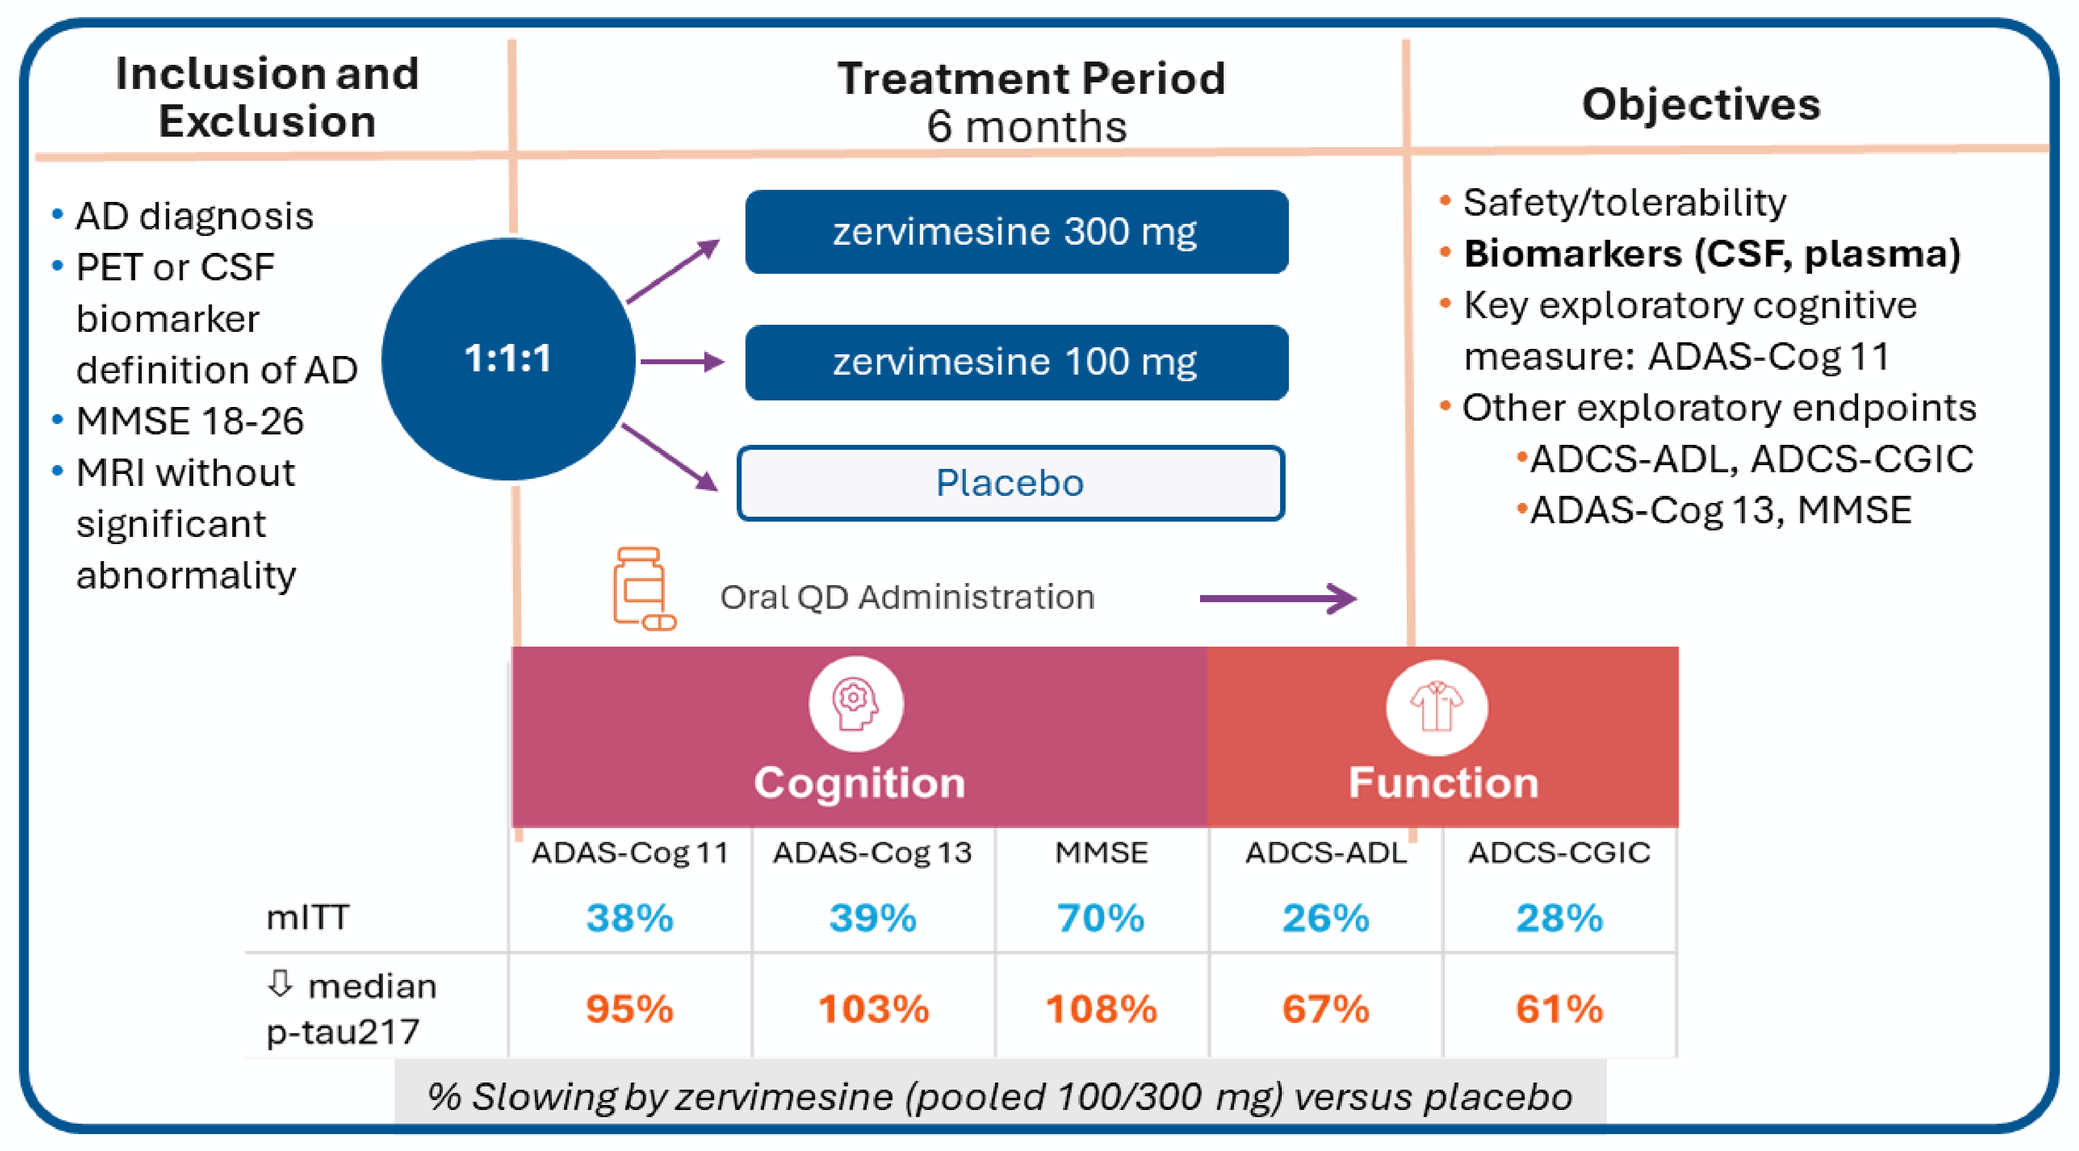
Inclusion and Exclusion
• AD diagnosis
• PET or CSF biomarker definition of AD
• MMSE 18-26
• MRI without significant abnormality
Treatment Period
6 months
1:1:1
zervimesine 300 mg
zervimesine 100 mg
Placebo
Oral QD Administration
Objectives
• Safety/tolerability
• Biomarkers (CSF, plasma)
• Key exploratory cognitive measure: ADAS-Cog 11
• Other exploratory endpoints
•ADCS-ADL, ADCS-CGIC
•ADAS-Cog 13, MMSE
Cognition
Function
ADAS-Cog 11
ADAS-Cog 13
MMSE
ADCS-ADL
ADCS-CGIC
mITT
38%
39%
70%
26%
28%
⇩ median p-tau217
95%
103%
108%
67%
61%
% Slowing by zervimesine (pooled 100/300 mg) versus placebo

Larger Biomarker Treatment Effect Observed in Below Median p-tau217 Group vs. mITT

Plasma biomarker (ng/L)		mITT (N*=150)	< median p-tau217 (N*=69)	> median p-tau217 (N*=69)
Aβ42	LS mean (SE)	-0.188 (0.2393)	-0.64 (0.326)	0.24 (0.322)
	95% CI	(-0.66, 0.29)	(-1.289, 0.018)	(-0.400, 0.885)
	p-value	0.4343	0.0565	0.4539
Aβ40	LS mean (SE)	-8.272 (5.0557)	-12.27 (7.296)	-1.07 (6.793)
	95% CI	(-18.28, 1.74)	(-26.869, 2.334)	(-14.653, 12.523)
	p-value	0.1044	0.0980	0.8759
GFAP	LS mean (SE)	-3.2 (10.53)	-28.35 (11.687)	23.07 (16.789)
	95% CI	(-24, 18)	(-51.769, -4.933)	(-10.544, 56.679)
	p-value	0.7593	0.0186	0.1748
NFL	LS mean (SE)	-1.44 (1.130)	-2.67 (1.503)	-0.22 (1.707)
	95% CI	(-3.7, 0.8)	(-5.681, 0.339)	(-3.637, 3.200)
	p-value	0.2057	0.0809	0.8987
p-tau217	LS mean (SE)	-0.053 (0.0569)	-0.10 (0.079)	0.02 (0.089)
	95% CI	(-0.17, 0.06)	(-0.264, 0.056)	(-0.154, 0.203)
	p-value	0.3581	0.1976	0.7884
Brain-derived tau	LS mean (SE)	-0.168 (0.0987)	-0.11 (0.089)	-0.19 (0.149)
	95% CI	(-0.36, 0.03)	(-0.293, 0.070)	(-0.491, 0.104)
	p-value	0.0907	0.2208	0.1987

zervimesine vs placebo; trend p≤0.1, significant p≤0.05

Proteomic Analysis Workflow

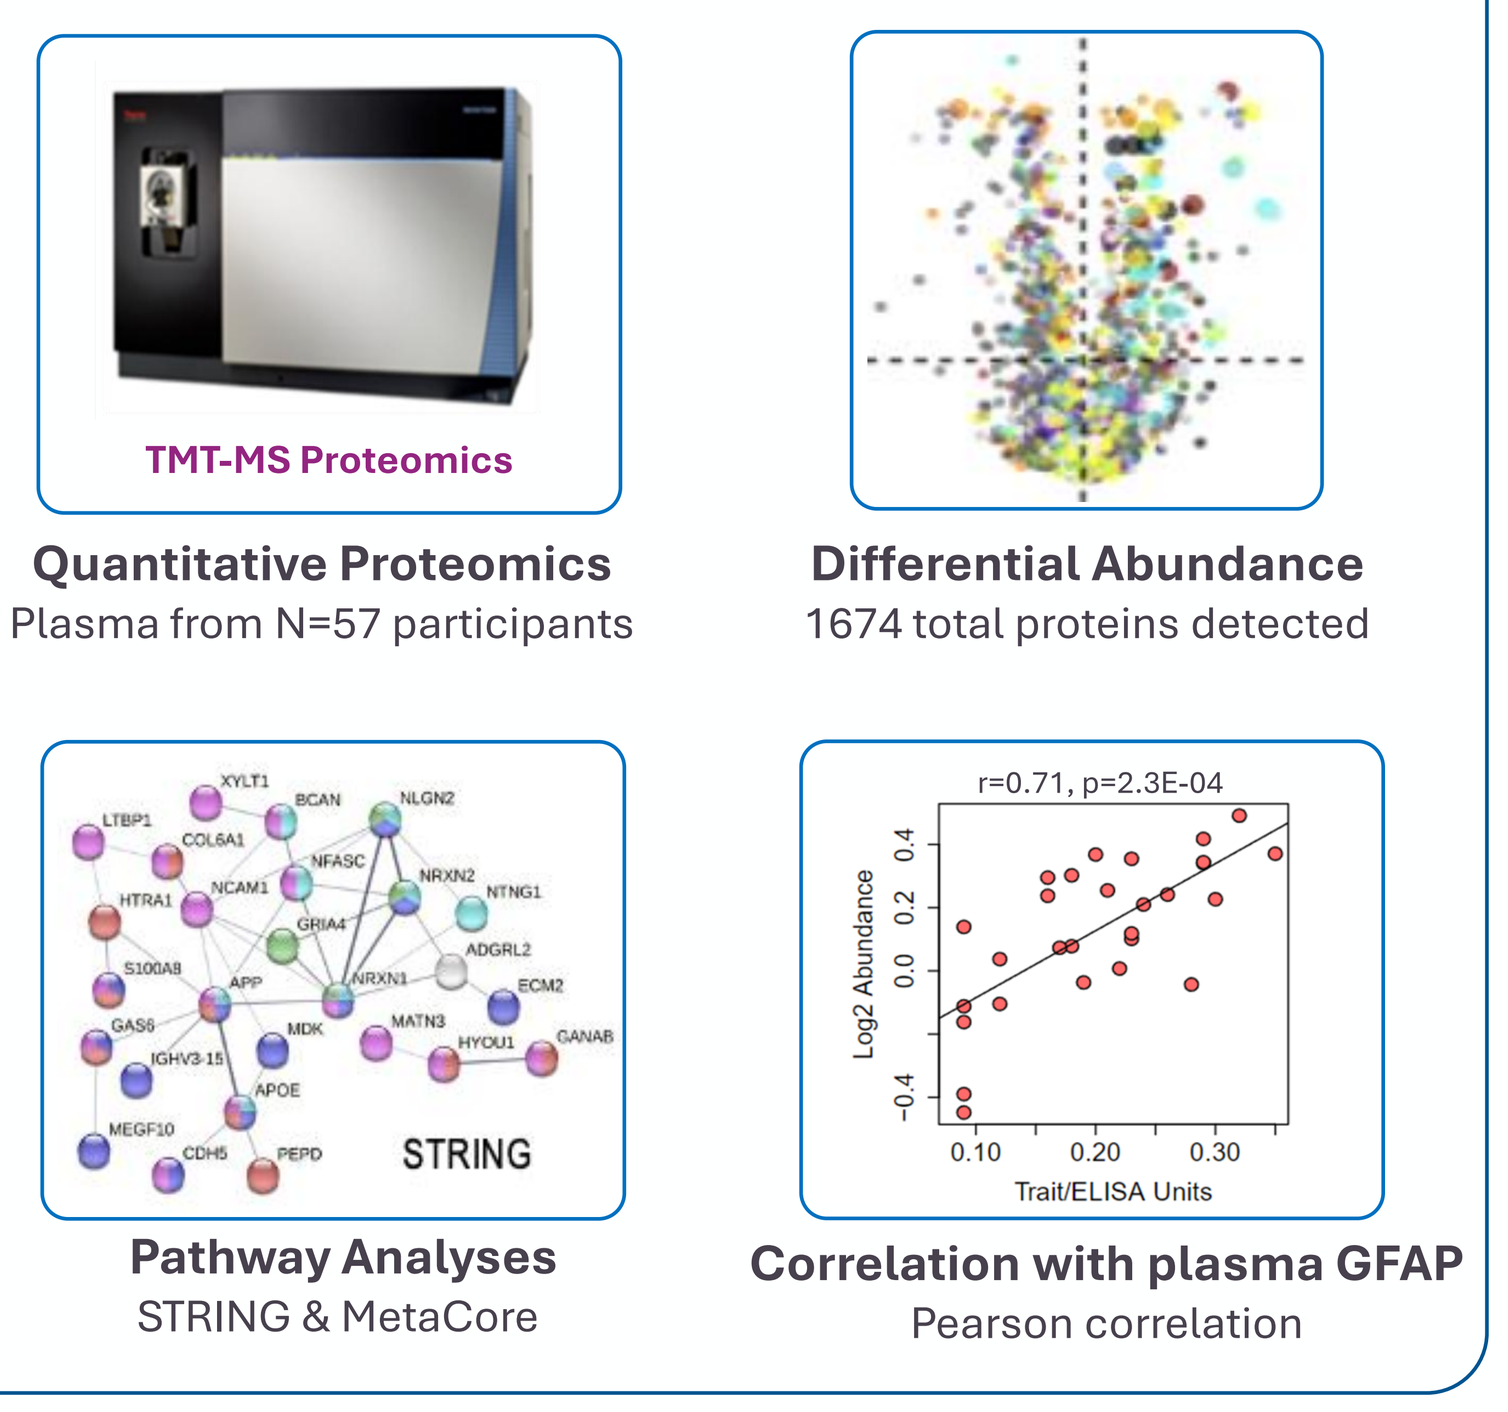
TMT-MS Proteomics
Quantitative Proteomics
Plasma from N=57 participants
Differential Abundance
1674 total proteins detected
STRING
Pathway Analyses
STRING & MetaCore
r=0.71, p=2.3E-04
Correlation with plasma GFAP
Pearson correlation

Results

Pharmacodynamic plasma biomarkers of zervimesine identified in proteomic analysis from mITT SHINE participants

A. Differentially Abundant Proteins (Zervimesine vs. Placebo; p≤0.05)

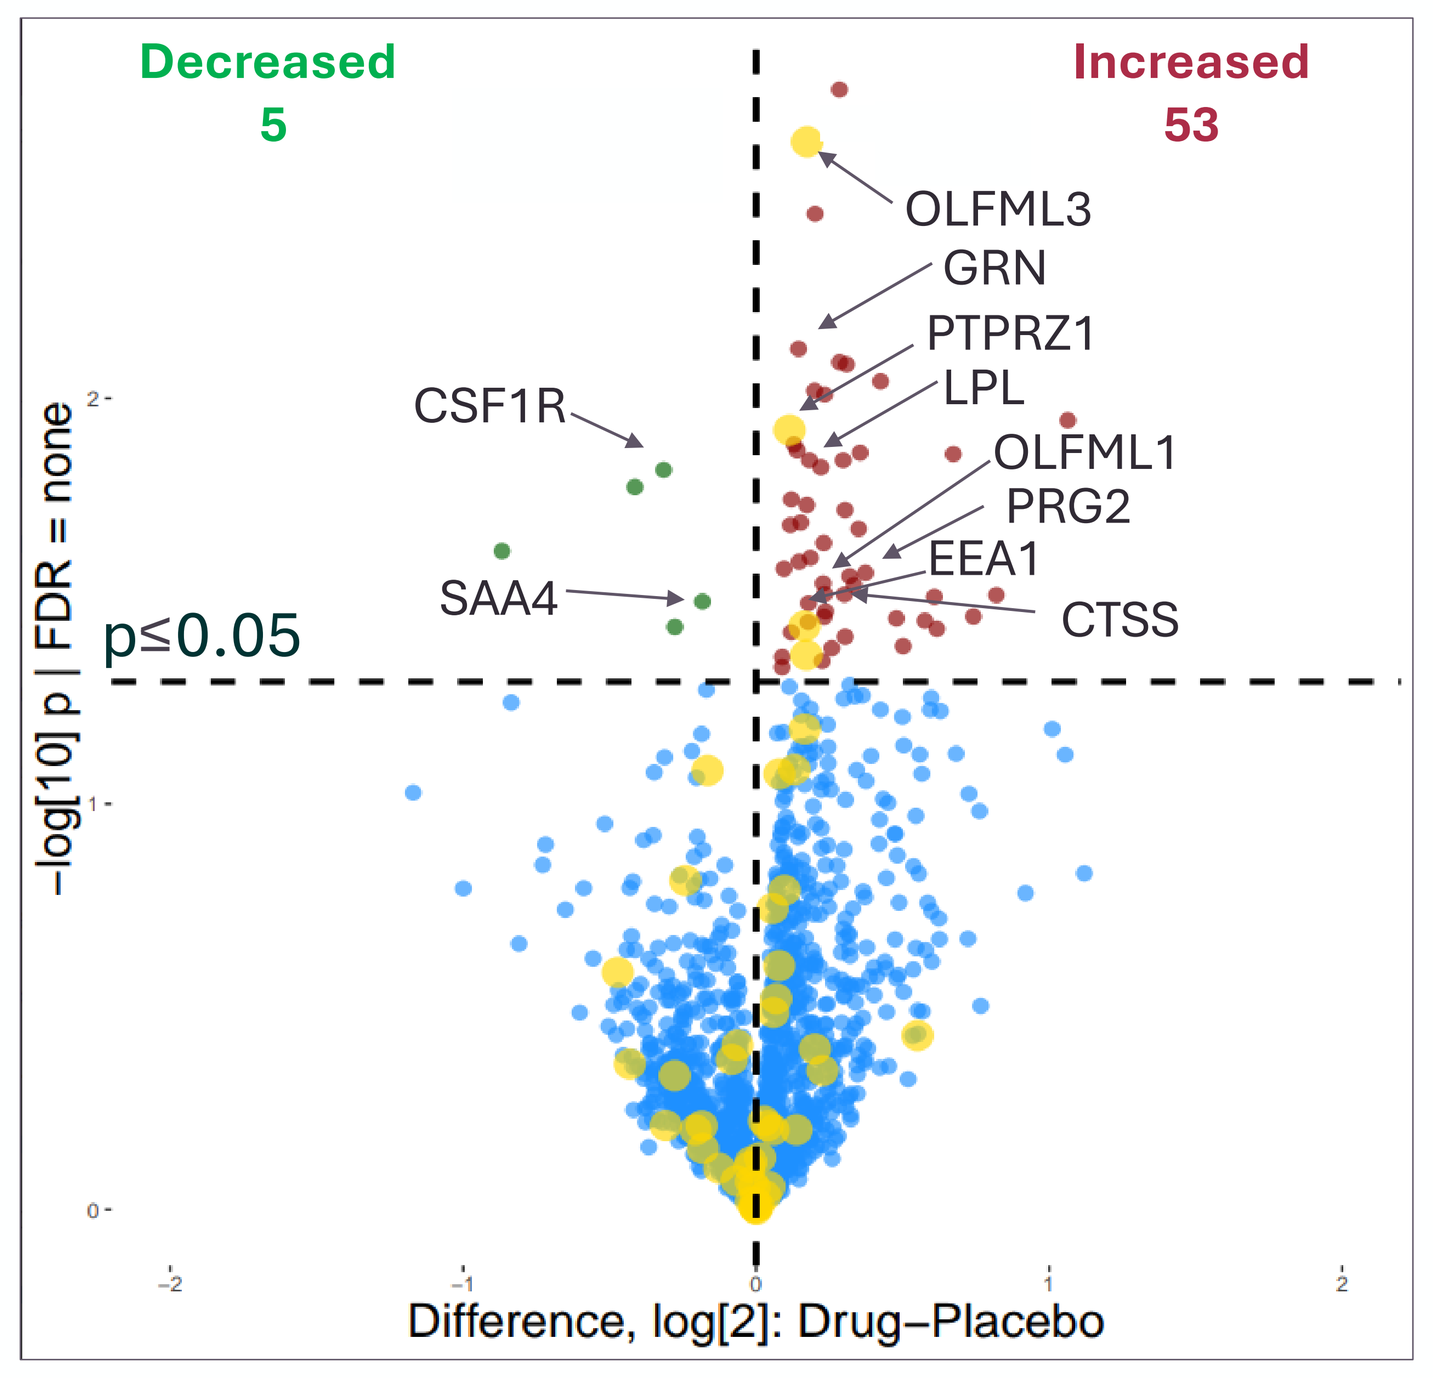
Decreased
5
Increased
53
OLFML3
GRN
PTPRZ1
LPL
OLFML1
PRG2
EEA1
CTSS
CSF1R
SAA4
p≤0.05
−log[10] p | FDR = none
Difference, log[2]: Drug−Placebo

B. Protein Abundance Variance Across Individual Samples (p≤0.05)

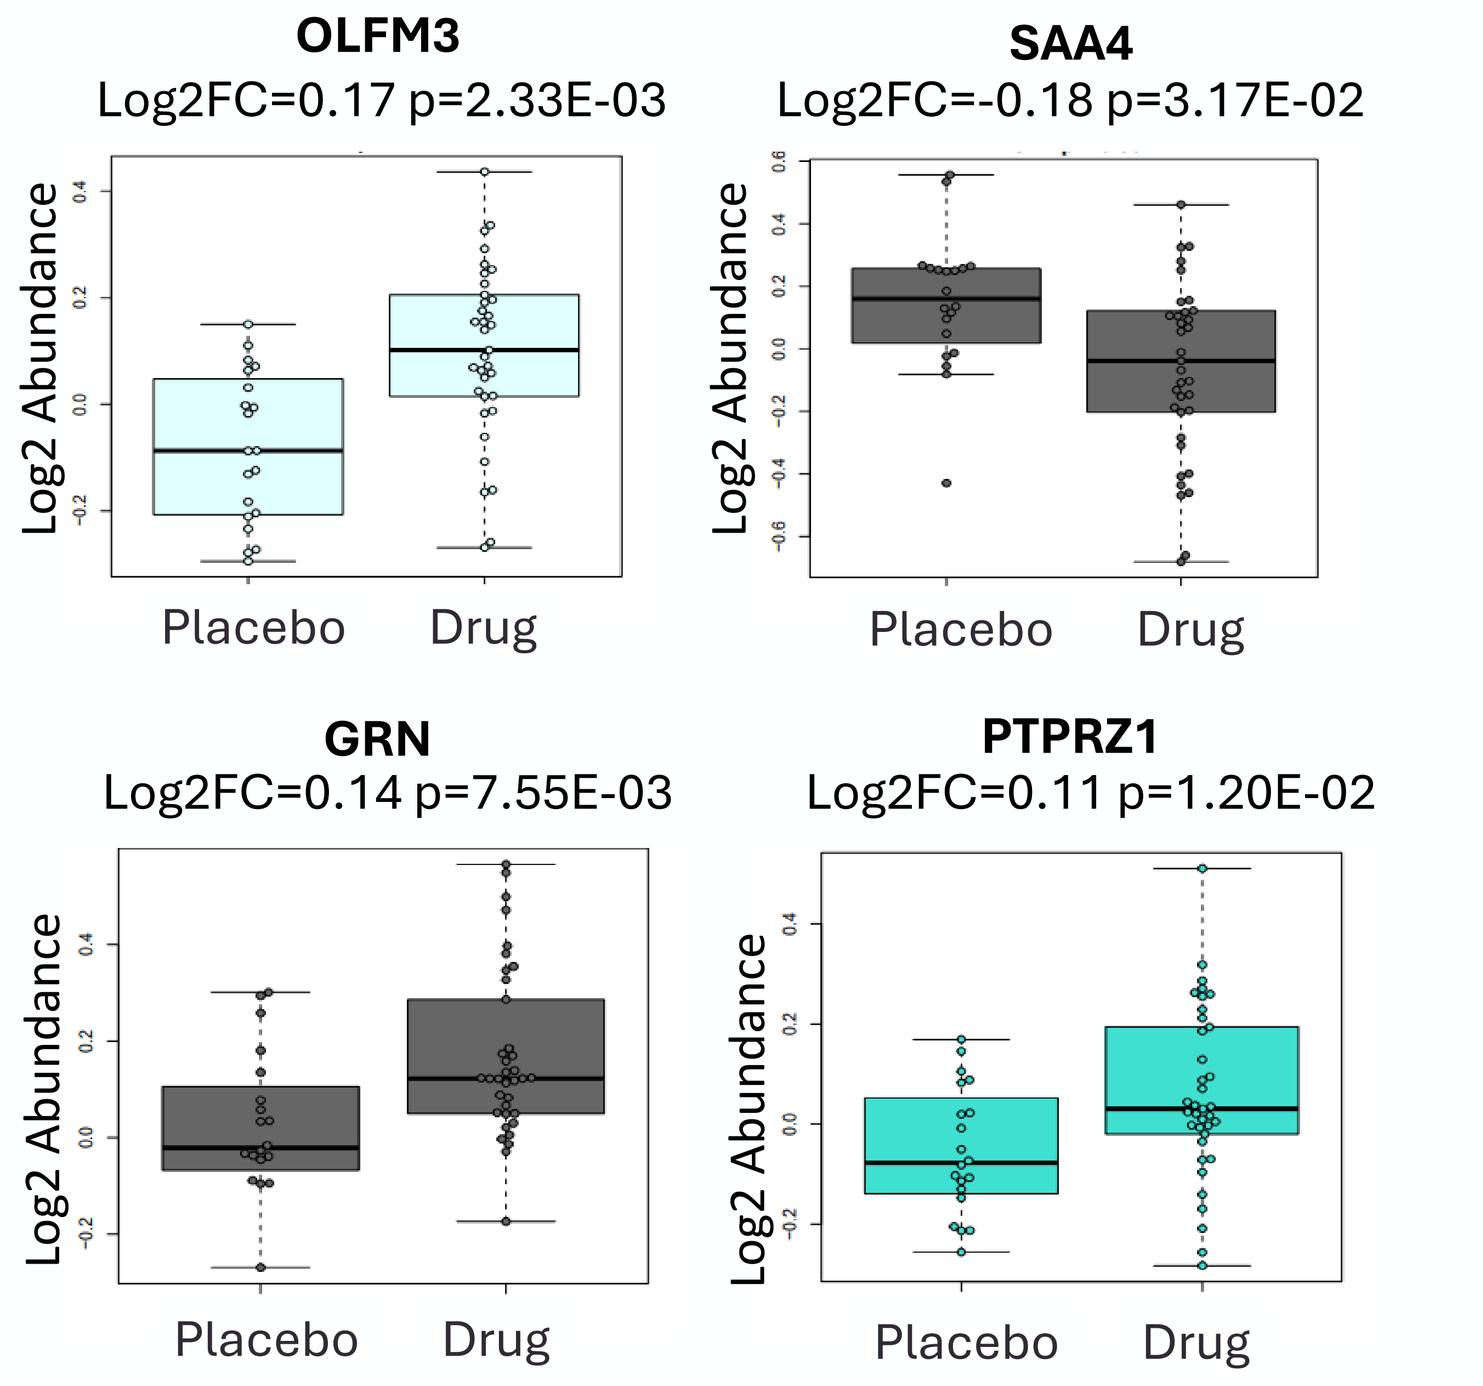
OLFM3
Log2FC=0.17 p=2.33E-03
SAA4
Log2FC=-0.18 p=3.17E-02
GRN
Log2FC=0.14 p=7.55E-03
PTPRZ1
Log2FC=0.11 p=1.20E-02
Log2 Abundance
Placebo
Drug

C. STRING Pathway Analysis (Zervimesine vs. Placebo, p≤0.05)

term ID	GO Component	Strength	FDR
GO:0005576	Extracellular region	0.61	1.86E-21
GO:0005615	Extracellular space	0.66	1.12E-18
GO:0062023	Collagen-containing extracellular matrix	1.16	1.01E-11
GO:1903561	**Extracellular vesicle**	0.68	4.99E-11
GO:0070062	**Extracellular exosome**	0.67	1.87E-10
GO:0031982	**Vesicle**	0.50	1.88E-09
GO:0030141	**Secretory granule**	0.88	1.96E-09
GO:0099503	**Secretory vesicle**	0.82	3.59E-09

D. MetaCore Pathway Analysis (Zervimesine vs. Placebo, p≤0.05)

Top MetaCore Pathway Maps	p-value
Fatty Acid Omega Oxidation	3.85E-03
Immune response_Antigen presentation by MHC class I, classical pathway	1.21E-02
Transcription_Effect of Folic acid on genome stability	1.48E-02
Development_Growth factors in regulation of oligodendrocyte progenitor cell proliferation	1.82E-02
Lipoprotein metabolism	1.87E-02
Transport_Macropinocytosis	3.66E-02

Figure 1. (A) Volcano plots to visualize directionality (58 proteins met significance criterion of p≤0.05 for zervimesine vs. placebo) in mITT participants. Each colored dot corresponds to a protein, larger yellow circles indicate AD priority biomarkers[6,7], proteins with known relationship to AD biology or immune response are indicated with black arrows. **(B)** Example of box plots of proteins of interest demonstrate the differential abundance and variance across individuals in mITT participants. Pathway analyses using both **(C)** STRING (sorted by FDR) and **(D)** MetaCore (version 24.4.71900) of differentially abundant proteins. Pathways related to AD or S2R biology are indicated in bold. Top Pathway maps exclude non-relevant tissues or diseases

Proteins shown in volcano plot: CSF1R, colony stimulating factor 1 receptor; CTSS, cathepsin S; EEA1, early endosome antigen 1; GRN, granulin precursor; LPL, lipoprotein lipase; OLFML1, olfactomedin like 1; OLFML3, olfactomedin like 3; PRG2, proteoglycan 2, pro eosinophil major basic protein; PTPRZ1, protein tyrosine phosphatase receptor type Z1; SAA4, serum amyloid A4.

PD biomarkers of zervimesine in the low p-tau217 group are associated with immune response, apoptosis and cell survival

A. Differentially Abundant Proteins (Zervimesine vs. Placebo; p≤0.05)

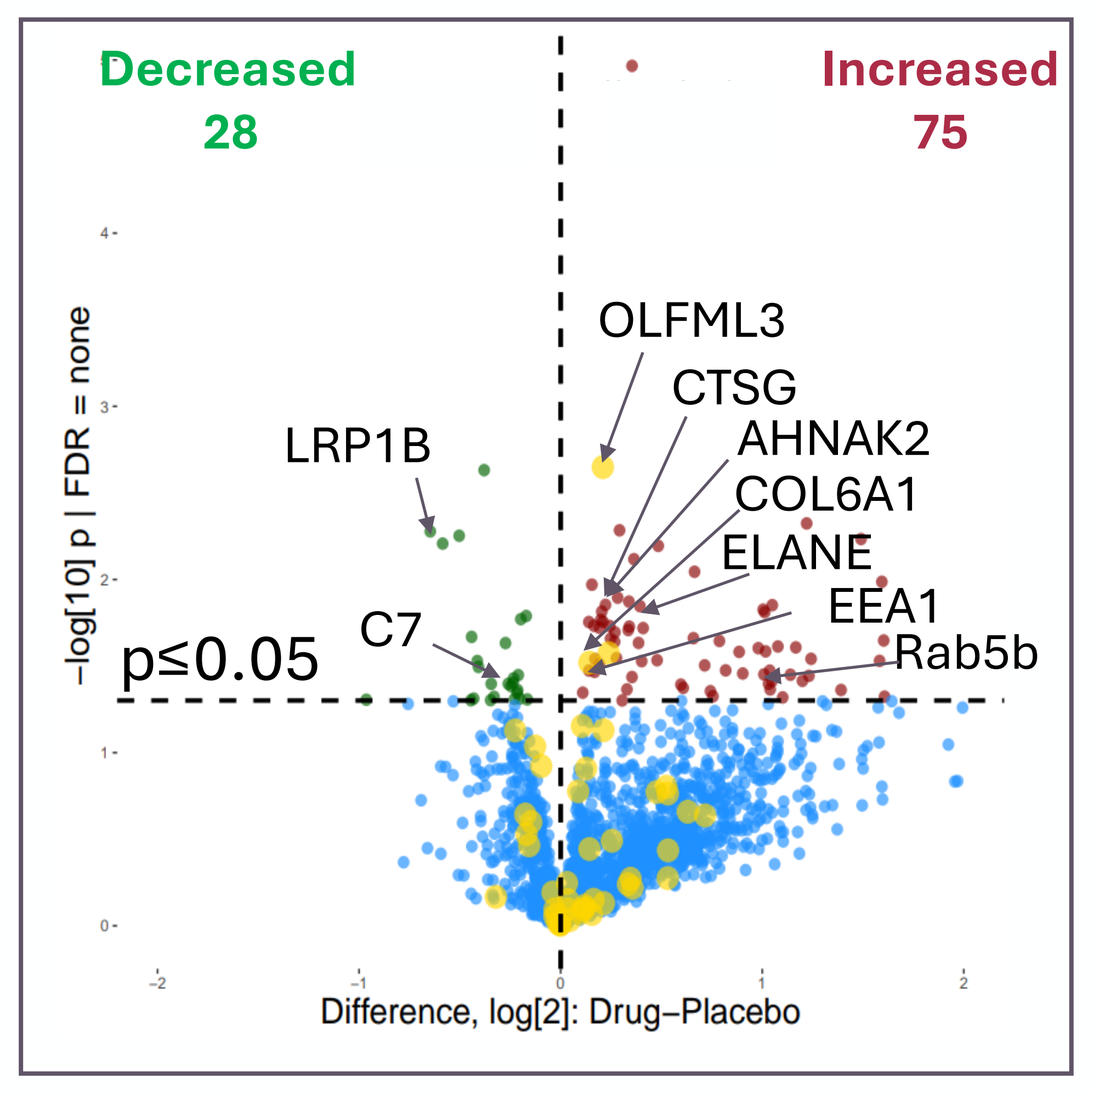
Decreased
28
Increased
75
OLFML3
CTSG
AHNAK2
COL6A1
ELANE
EEA1
Rab5b
LRP1B
C7
p≤0.05
−log[10] p | FDR = none
Difference, log[2]: Drug−Placebo

B. MetaCore Pathway Analysis (Zervimesine vs. Placebo, p≤0.05)

Top MetaCore Pathway Maps	p-value
Development_Regulation of endothelial progenitor cell differentiation from adult stem cells	7.90E-06
Development_Role of G-CSF in hematopoietic stem cell mobilization	1.17E-04
LRRK2 in neurons in Parkinson's disease	4.60E-04
Apoptosis and survival_Granzyme A signaling	8.76E-04
Signal transduction_CXCR4 signaling via MAPKs cascades	1.85E-03
Signal transduction_Glucocorticoid receptor signaling	1.95E-03
Cytoskeleton remodeling_Keratin filaments	2.64E-03
Apoptosis and survival_FasL(TNFSF6)/ FasR(CD95)-induced cell death	4.43E-03
Immune response_IL-6 signaling via MEK/ERK and PI3K/AKT cascades	4.78E-03
Transport_Induction of Macropinocytosis	5.15E-03
Main pathways of Schwann cells transformation in neurofibromatosis type 1	5.95E-03
Gamma-secretase proteolytic targets	5.95E-03
G-protein signaling_Ras family GTPases in kinase cascades	6.32E-03
Immune response_Role of HMGB1 in dendritic cell maturation and migration	6.32E-03

Figure 2. (A) Volcano plots to visualize directionality (103 proteins met significance criterion of p≤0.05 for zervimesine vs. placebo) in low p-Tau217 participants. Each colored dot corresponds to a protein, larger yellow circles indicate AD priority biomarkers[6,7], proteins with known relationship to AD biology or immune system are indicated with black arrows. **(B)** Pathway analyses using MetaCore (version 24.4.71900) (sorted by p-value) of differentially abundant proteins at p≤0.05 in plasma proteome in the low p-tau217 group. Pathways of interest related to AD or neuronal biology are indicated in bold. Top Pathway maps exclude non-relevant tissues or diseases.

Proteins shown in volcano plot: C7, complement 7; COL6A1, collagen type V1 alpha 1 chain; CTSG, cathepsin G; EEA1, early endosome antigen 1; LRP1B, LDL receptor related protein 1B; OLFML3, olfactomedin like 3; Rab5b, member RAS oncogene family; AHNAK2, AHNAK nucleoprotein 2; ELANE, elastase, neutrophil expressed.

Pharmacodynamic biomarkers of zervimesine that correlate with GFAP levels identified in low p-tau217 group

A.

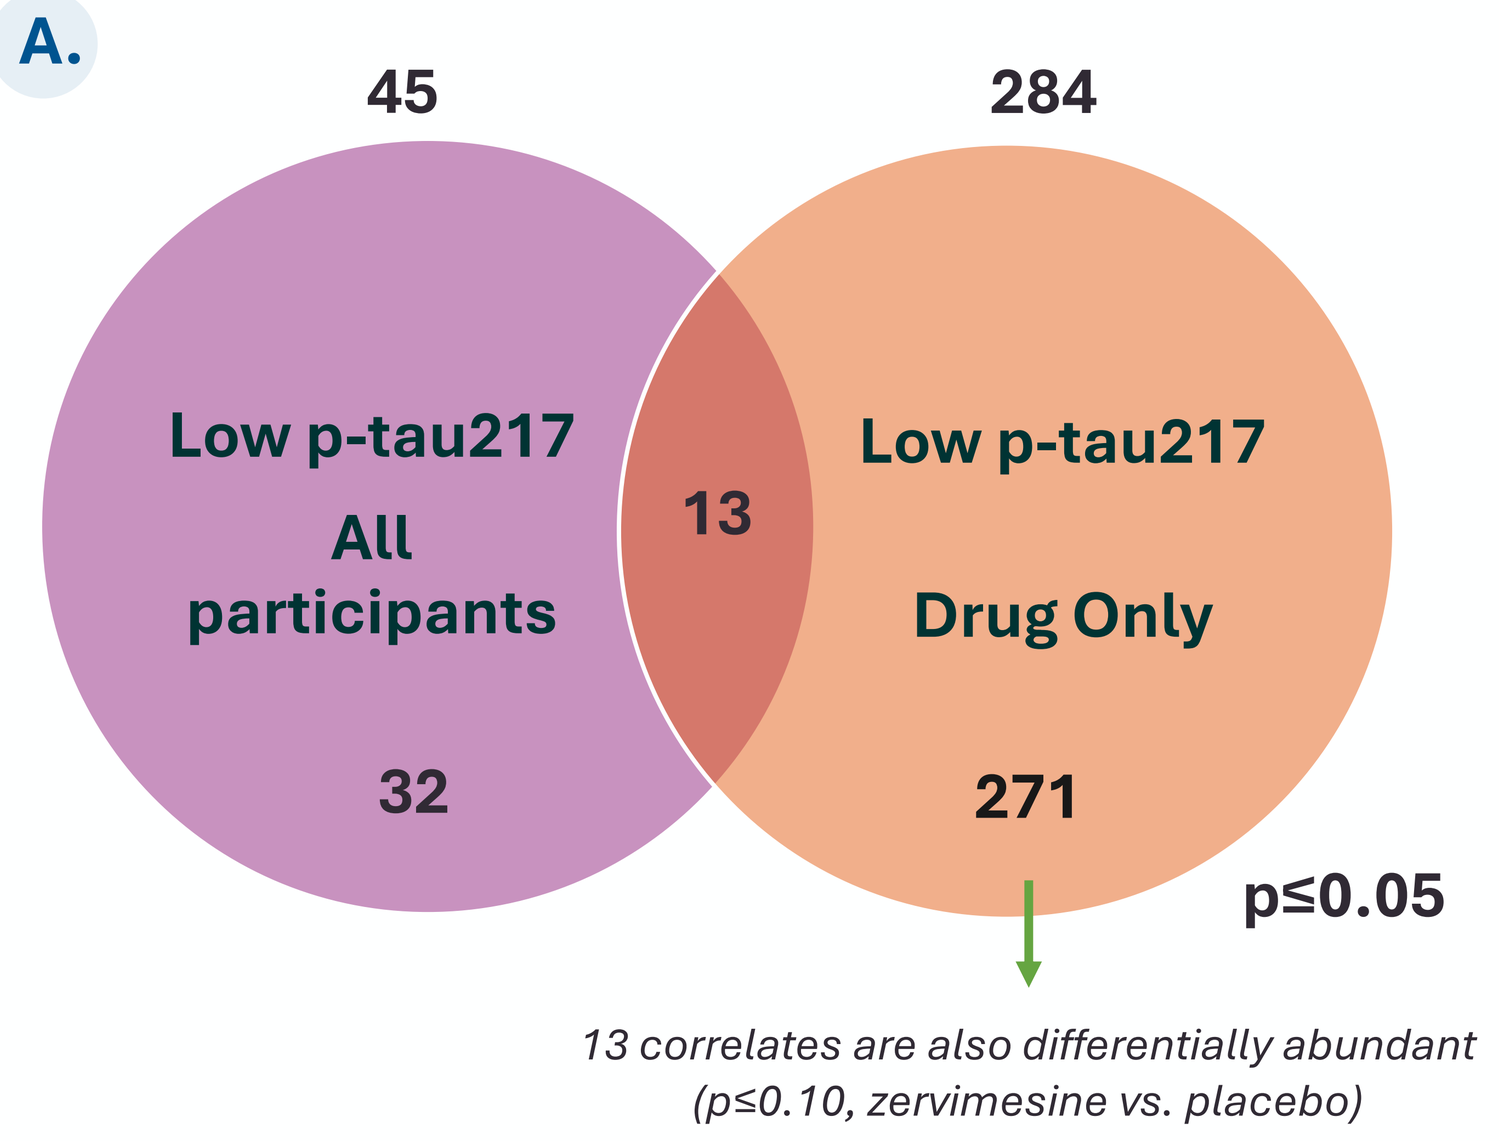
45
284
Low p-tau217
All participants
32
13
Low p-tau217
Drug Only
271
p≤0.05
13 correlates are also differentially abundant
(p≤0.10, zervimesine vs. placebo)

B.

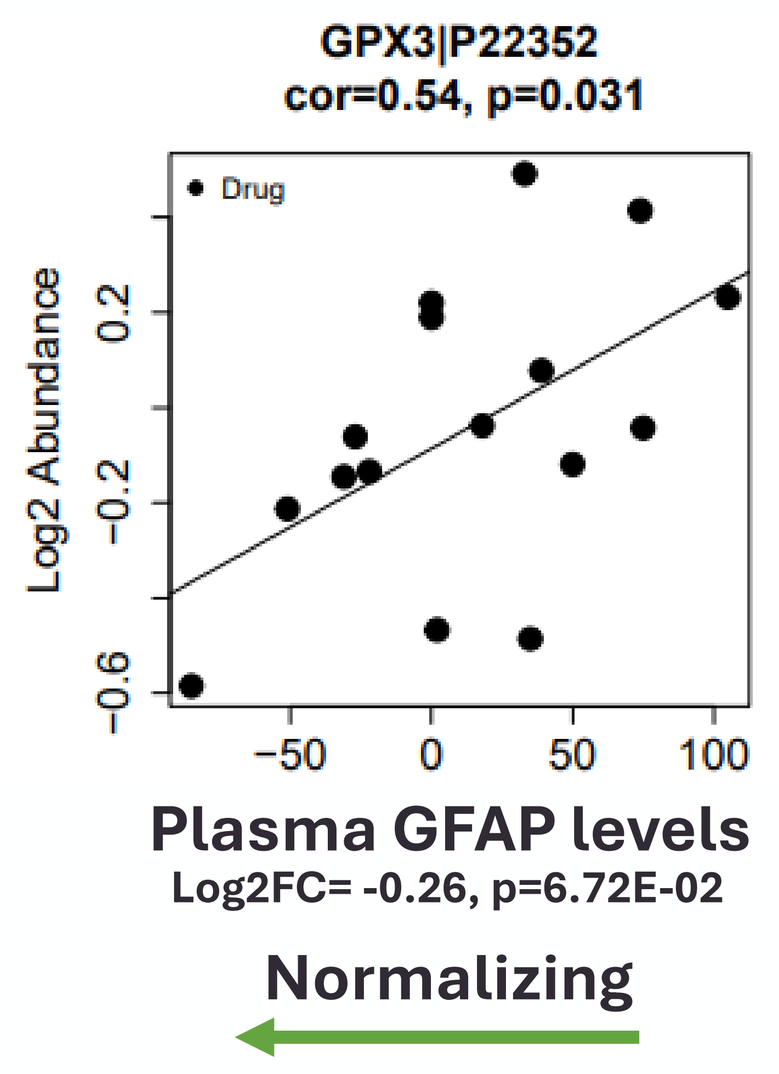
GPX3|P22352
cor=0.54, p=0.031
Drug
Log2 Abundance
Plasma GFAP levels
Log2FC= -0.26, p=6.72E-02
Normalizing

Figure 3. (A) Venn diagram to show common correlates (p≤0.05). Plasma proteins with change from baseline (CFB) abundances correlated with changes in plasma GFAP levels (correlates) identified in "All participants" and "Drug Only" groups in the low p-tau217 group. **(B)** Representative scatter plot to show favorable correlation of GPX3 with plasma GFAP levels in individual zervimesine-treated patients.

Drug-selective proteins correlated with plasma GFAP involved in pathways related to S2R mechanism and immune response

A. STRING Pathway Analysis of "Drug-Selective" Correlates with Plasma GFAP Levels

term ID	Reactome	Strength	FDR
HSA-109582	Hemostasis	0.83	6.21E-25
HSA-194315	Signaling by Rho GTPases	0.80	6.46E-25
HSA-5653656	**Vesicle-mediated transport**	0.79	1.10E-23
HSA-199991	**Membrane Trafficking**	0.78	1.35E-21
HSA-76002	Platelet activation, signaling and aggregation	1.01	1.35E-21
HSA-168256	**Immune System**	0.51	1.77E-20
HSA-162582	Signal Transduction	0.46	2.07E-20
HSA-6798695	Neutrophil degranulation	0.81	5.80E-18
HSA-195258	RHO GTPase Effectors	0.92	2.00E-17
HSA-168249	**Innate Immune System**	0.61	6.62E-17
HSA-114608	Platelet degranulation	1.13	6.90E-16
HSA-1643685	Disease	0.49	5.60E-15
HSA-9012999	RHO GTPase cycle	0.76	4.66E-14
HSA-422475	**Axon guidance**	0.71	8.86E-14
HSA-5663205	Infectious disease	0.59	2.35E-13

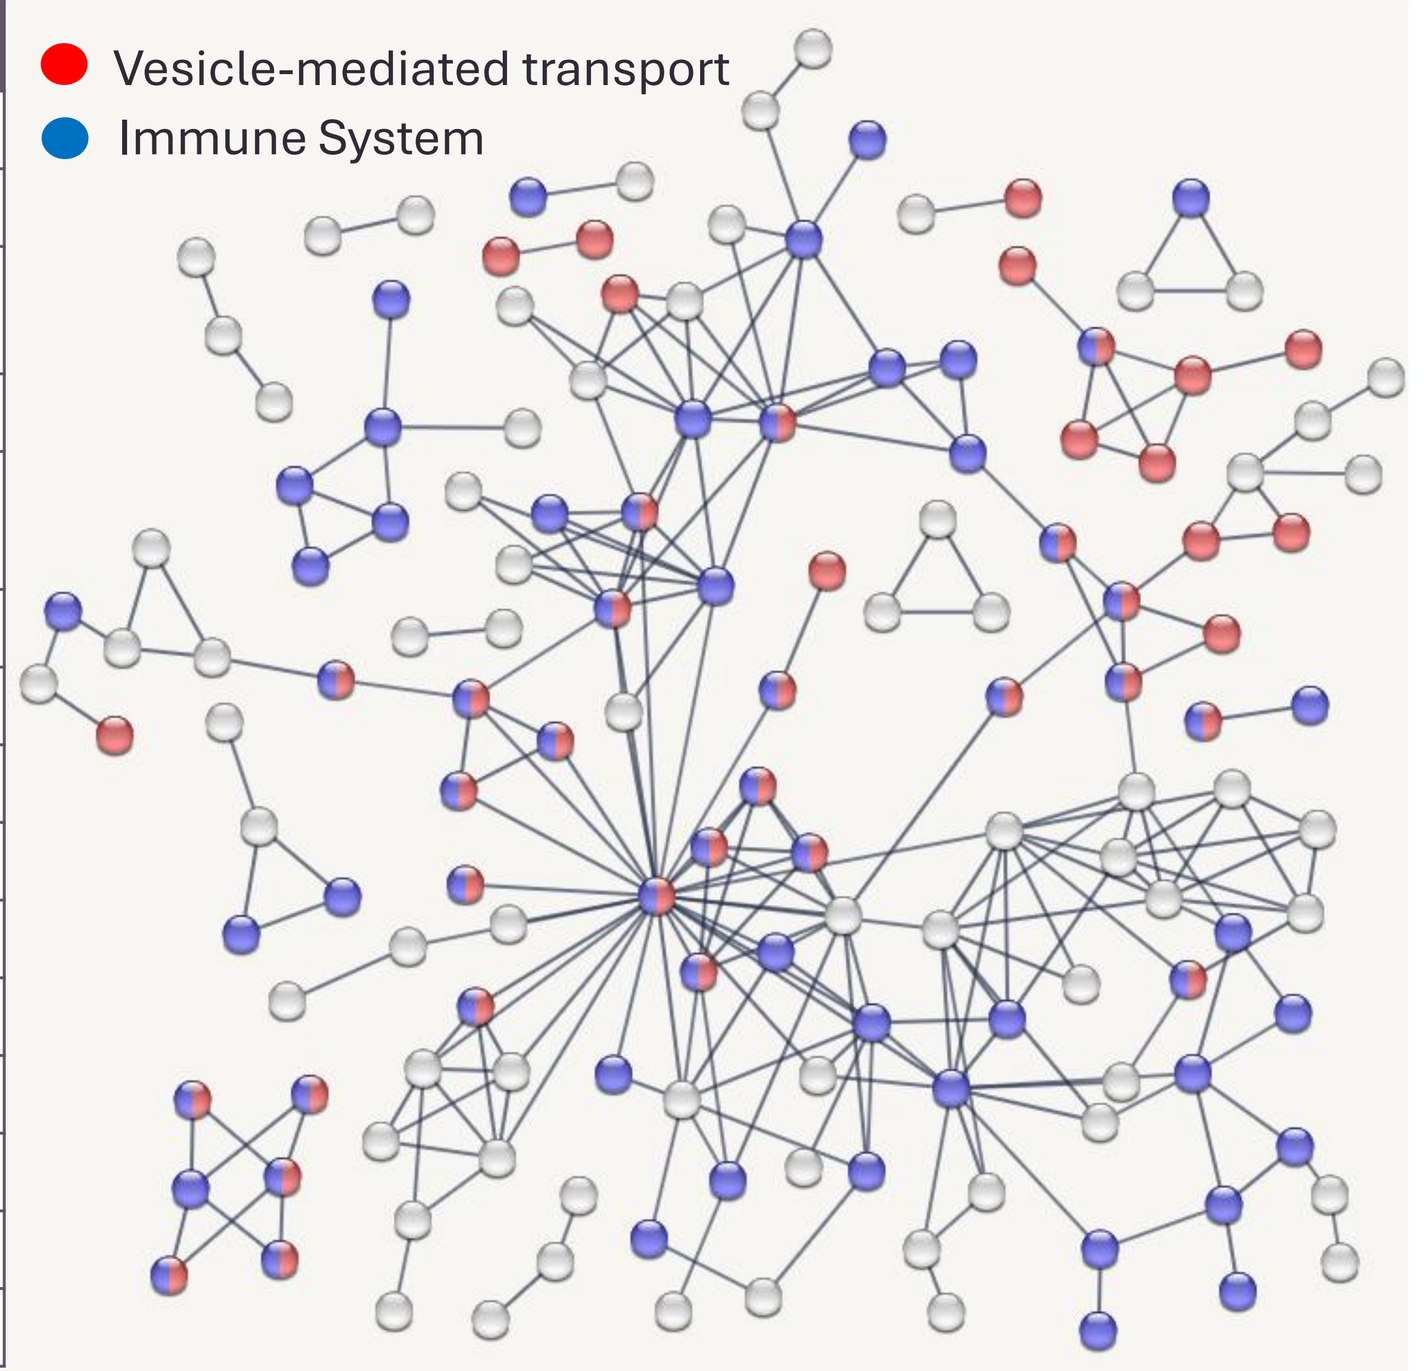
Vesicle-mediated transport
Immune System

B. MetaCore Pathway Analysis of "Drug-Selective" Correlates with Plasma GFAP Levels

Top MetaCore Pathway Maps	p-value
Immune response_Antigen presentation by MHC class I, classical pathway	4.26E-08
Cytoskeleton remodeling_Regulation of actin cytoskeleton nucleation and polymerization by Rho GTPases	2.70E-06
Immune response_Down-regulation of mast cell functions through ITIM-containing inhibitory receptors	9.79E-06
CFTR folding and maturation (normal and cystic fibrosis)	1.55E-05
HSP70 and HSP40-dependent folding in Huntington's disease	1.92E-05
Immune response_Antigen presentation by MHC class II	3.39E-05
Immune response_Antigen presentation by MHC class I: cross-presentation	6.18E-05
Immune response_Immunological synapse formation	9.96E-05
Cell adhesion_Integrin inside-out signaling in neutrophils	1.00E-04
G-protein signaling_TC21 regulation pathway	3.41E-04
Signal transduction_Thrombospondin 1 signaling	4.11E-04
Cell adhesion_Role of tetraspanins in the integrin-mediated cell adhesion	5.80E-04
Immune response_Inhibition of mast cell functions by Fc gamma RII beta	7.00E-04
Development_MAG, Reticulon 4 and OMgp in inhibition of neurite outgrowth	7.29E-04

Figure 4. (A) STRING Reactome pathways (sorted by FDR) and PPI map of the significant (p≤0.05) "Drug Selective" 271 correlates in the low p-tau217 group (confidence threshold=0.9; unconnected proteins hidden for visualization). Colored nodes refer to Reactome "Vesicle-mediated transport" (red), "Immune System" (blue). **(B)** Top-ranked pathways (sorted by p-value) identified by MetaCore (version 25.2.72100). Pathways of interest related to immune response or neuronal biology are indicated in bold.

Conclusions

- **Plasma PD biomarkers of zervimesine (CT1812) were identified from SHINE, in the mITT population and in the p-tau217 biomarker-defined subgroup**
- **The set of proteomic protein correlates of the canonical neuroinflammation biomarker GFAP may represent an inflammatory signature tied to the favorable dampening of neuroinflammation (GFAP) seen in zervimesine-treated participants**
- **These results support a role for zervimesine in decreasing neuroinflammation in a biomarker-defined patient population, and paired with the favorable effect on cognition, supports the continued clinical development of zervimesine for AD**

Other Presentations by Cognition Therapeutics at AAIC 2025

Poster #102120, Sunday July 27: Exploratory CSF proteomic analysis of a pre-specified p-tau217 subgroup from the SHINE clinical trial identifies biomarkers correlated with cognitive improvement in Alzheimer's disease patients treated with zervimesine

Poster #106858, Sunday July 27: Zervimesine (CT1812) Treatment Benefits Patients with Lower Baseline Plasma p-tau217 Across the Mild-to-Moderate AD Spectrum

Featured Research Session, Tuesday July 29, 8:00-8:45AM : Baseline Characteristics and Results of the Phase 2 COG1201 SHIMMER Study of zervimesine (CT1812)

This work was supported by funding from the National Institute on Aging (RF1AG057553 and R01AG058660)

CT1812 is an investigational therapeutic that has not been approved for any use by the US Food and Drug Administration. The USAN Council has adopted zervimesine as the United States Adopted Name (USAN) for CT1812.

References: 1. Clinical trials NCT03493282, NCT03507790, 2. Clinical trial NCT05225415, 3. Lizama BN, et al. *Int J Mol Sci*. 2023 Mar 26;24(7):6251., 4. Izzo NJ et al., *PLoS One*. 2014 Nov 12;9(11):e111898. 5. Hamby ME et al., AD/PD 2025 Podium Presentation, ID 3184. 6.Higginbotham L et al. *Sci Adv*. 2020 Oct 21;6(43):eaaz9360. 7. Johnson ECB et al. *Nat Neurosci*. 2022 Feb;25(2):213-225.

CONTACT:
mhamby@cogrx.com